HOW EFFICIENCY FOR IDENTIFYING OBJECTS IMPROVES WITH AGE

Asmita Mishra, Gunjan Baweja, Marialuisa Martelli, Isabelle Chen, Jeremy Fox, Najib Majaj, and Denis Pelli, Psychology & Neural Science, NYU

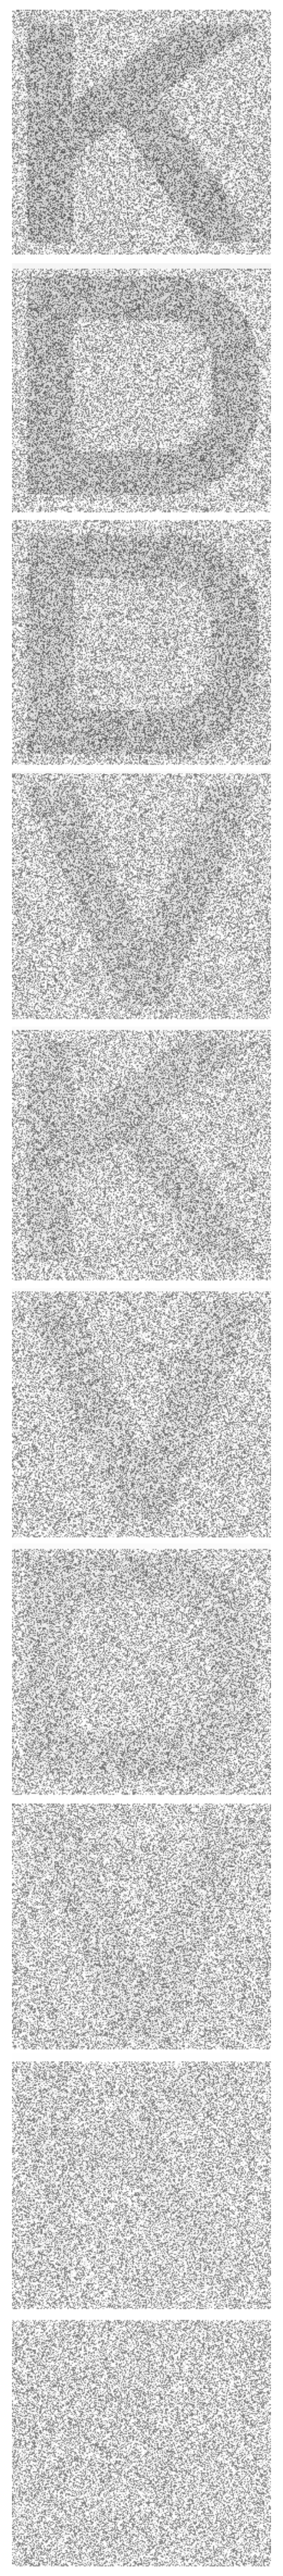

Letters

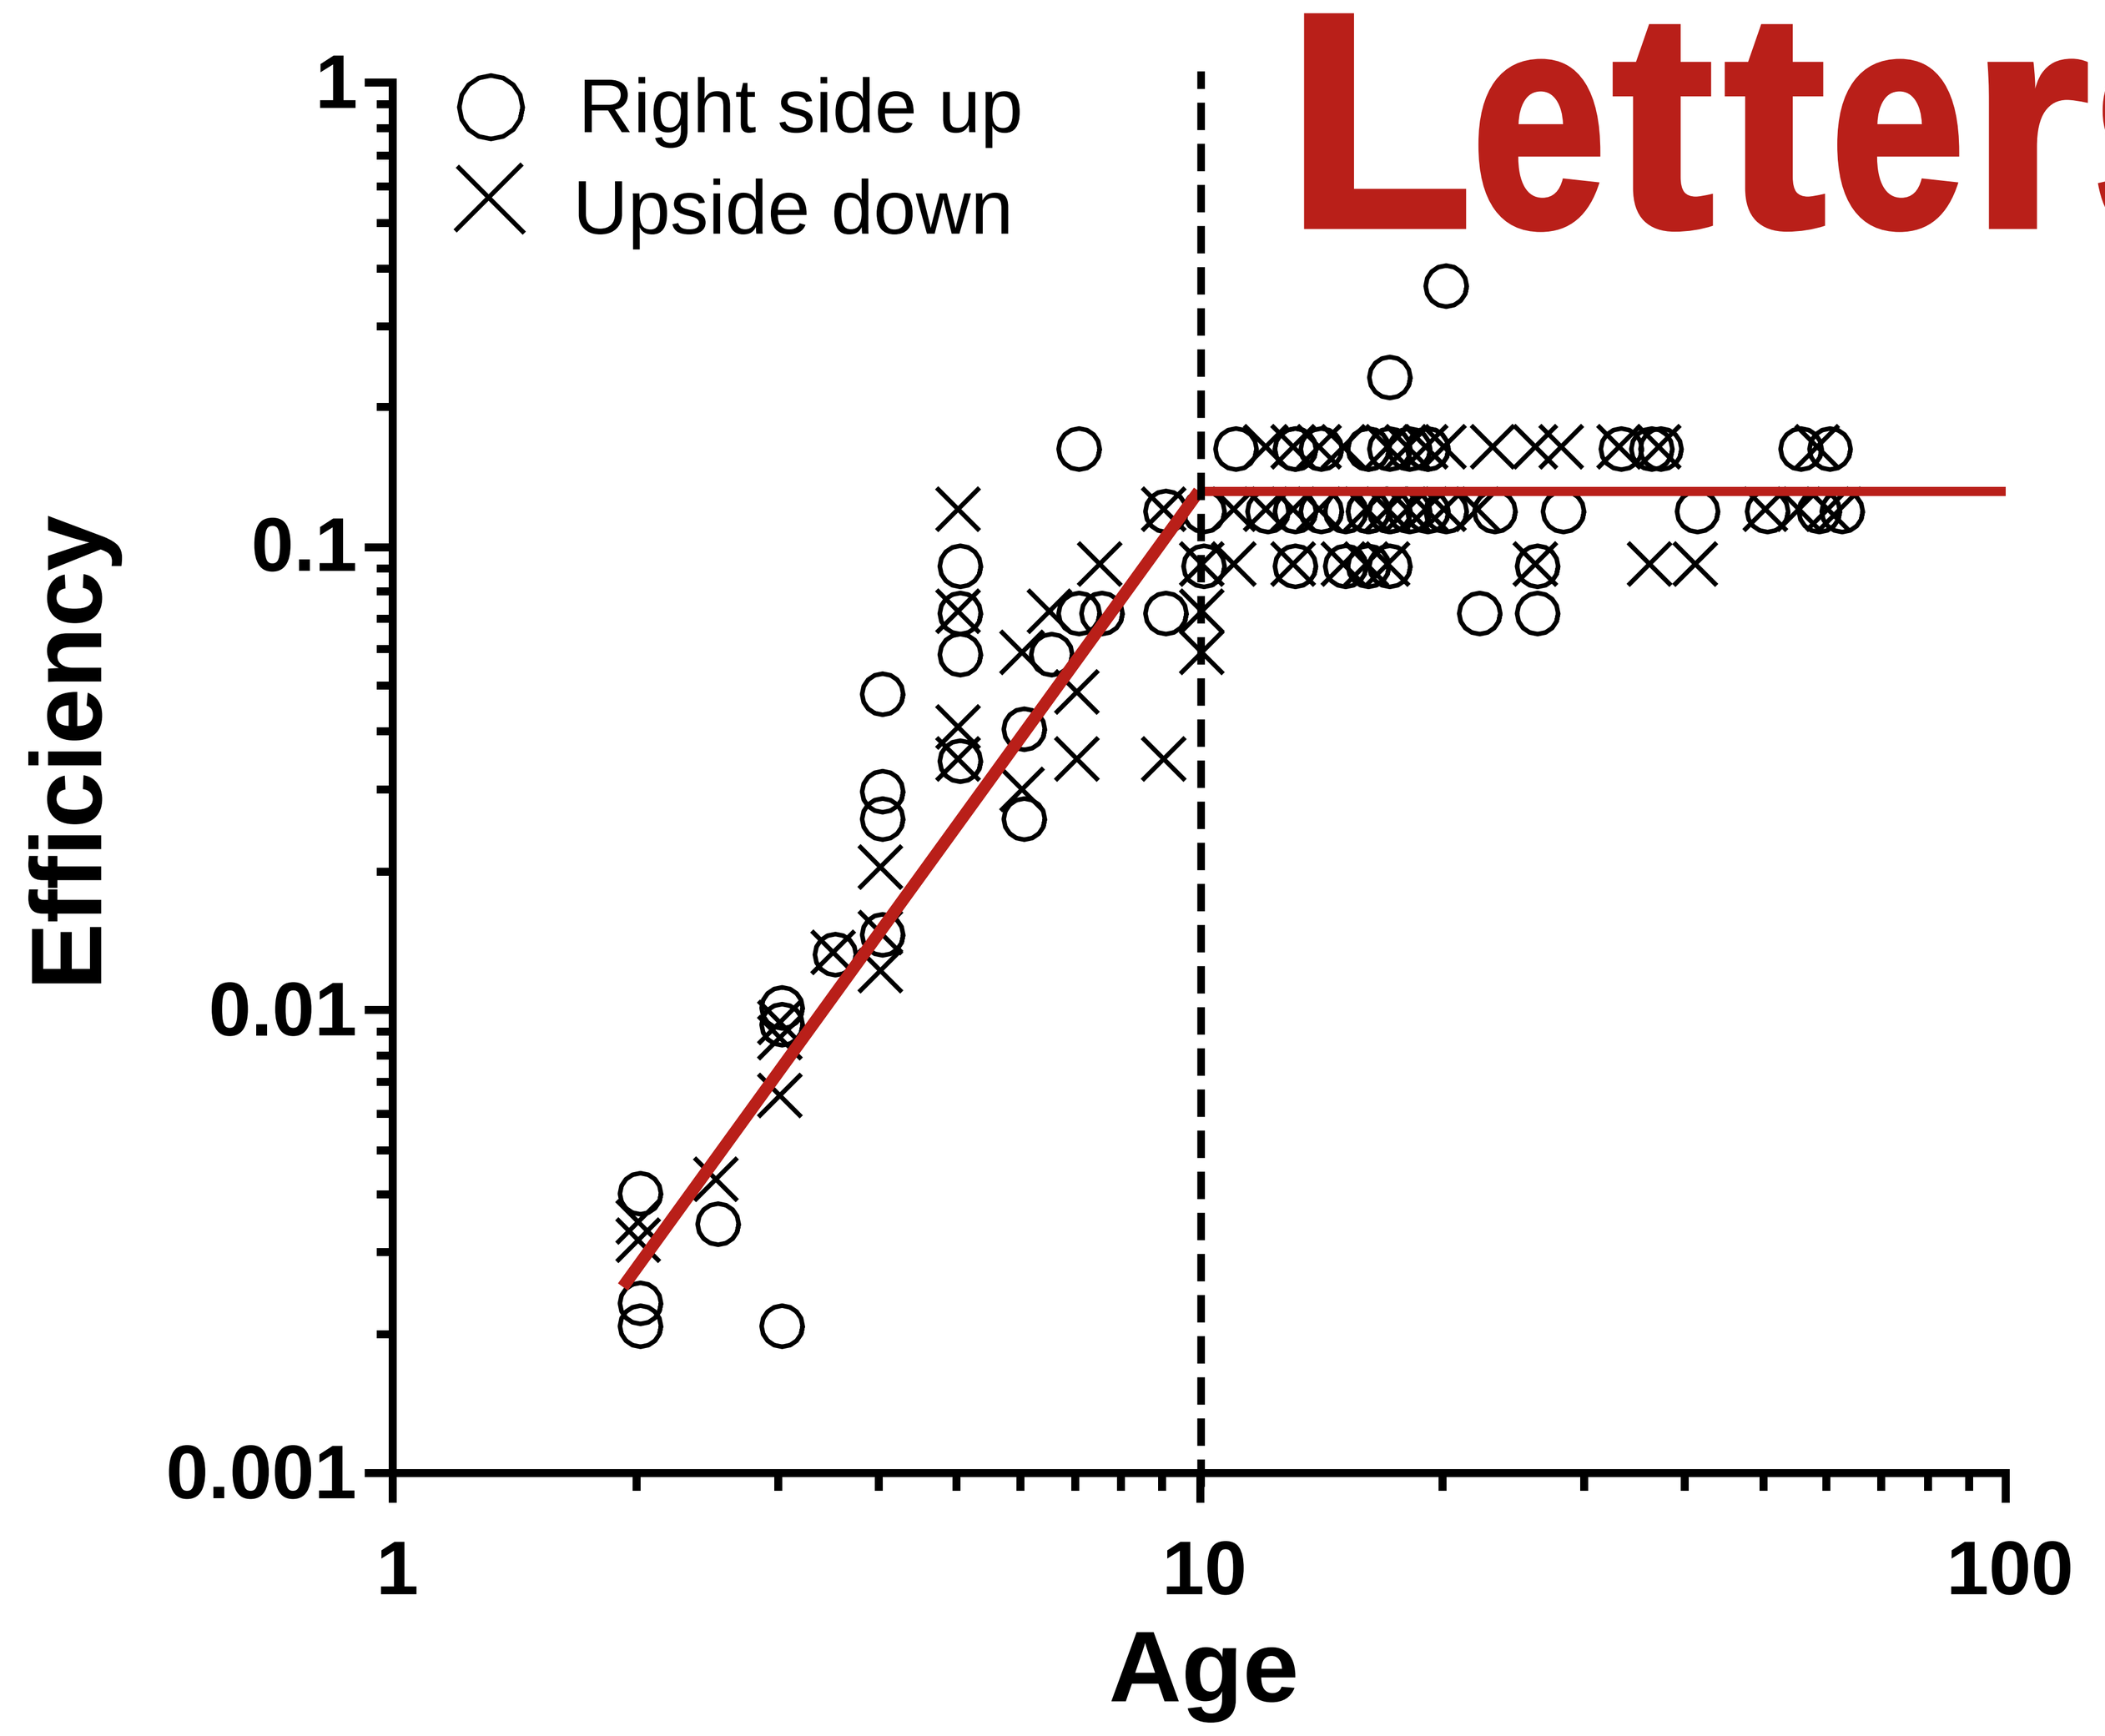
Right side up
Upside down
Efficiency
1
0.1
0.01
0.001
1
10
100
Age

INTRODUCTION: It has been proposed that object recognition can be attributed to separate category-specific recognition systems. However, letters, objects, and faces may differ in the degree of discriminability between the stimuli within a set. Efficiency (E_{ideal}/E^{+}) is an absolute measure of performance that allows for comparisons between different tasks. Efficiency gives the fraction of energy used by the human observer to do the task that is needed by the ideal observer. The ideal observer knows the possible signals and, given the noisy stimulus, chooses the likeliest possibility.

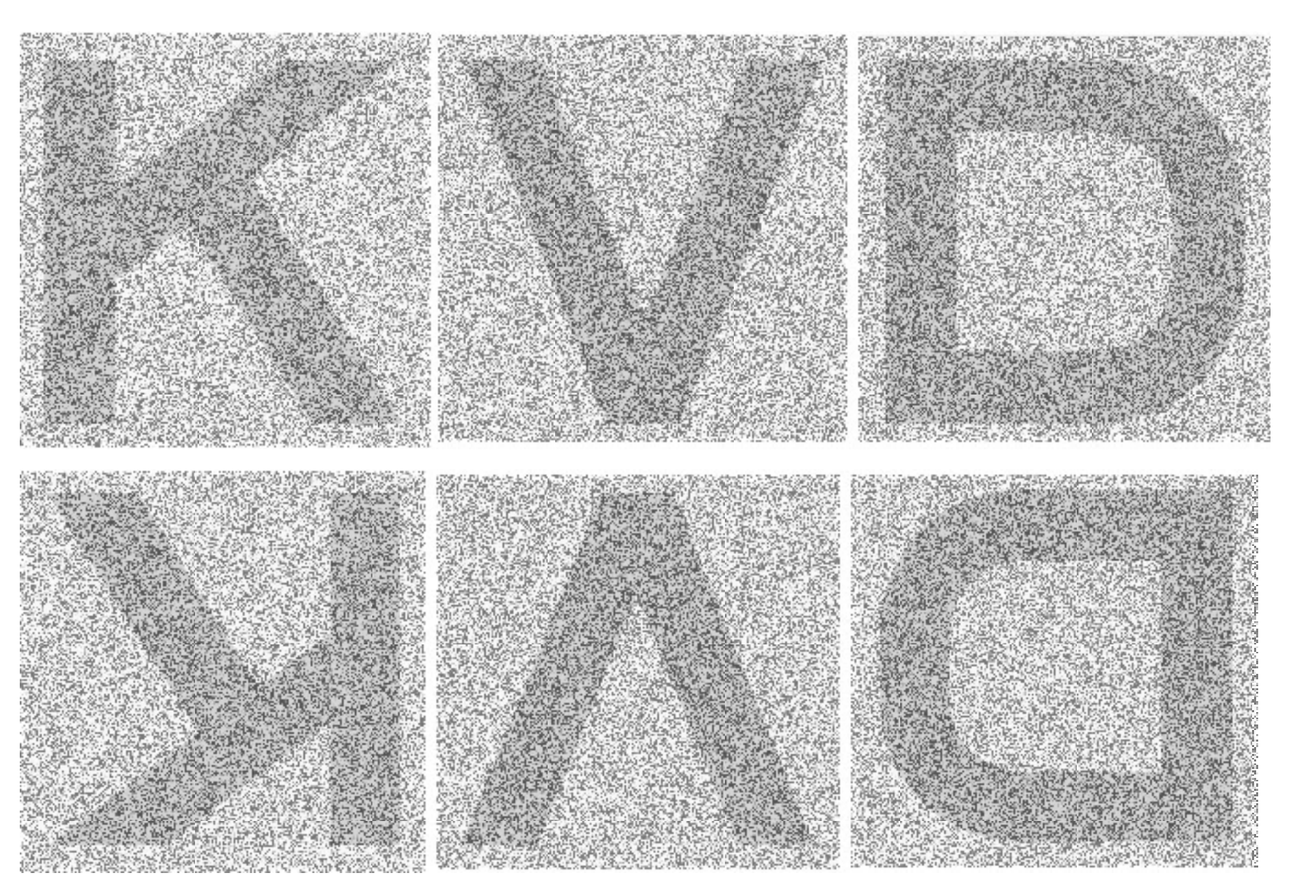

The vertical eye chart to the left uses these letters to measure your threshold.

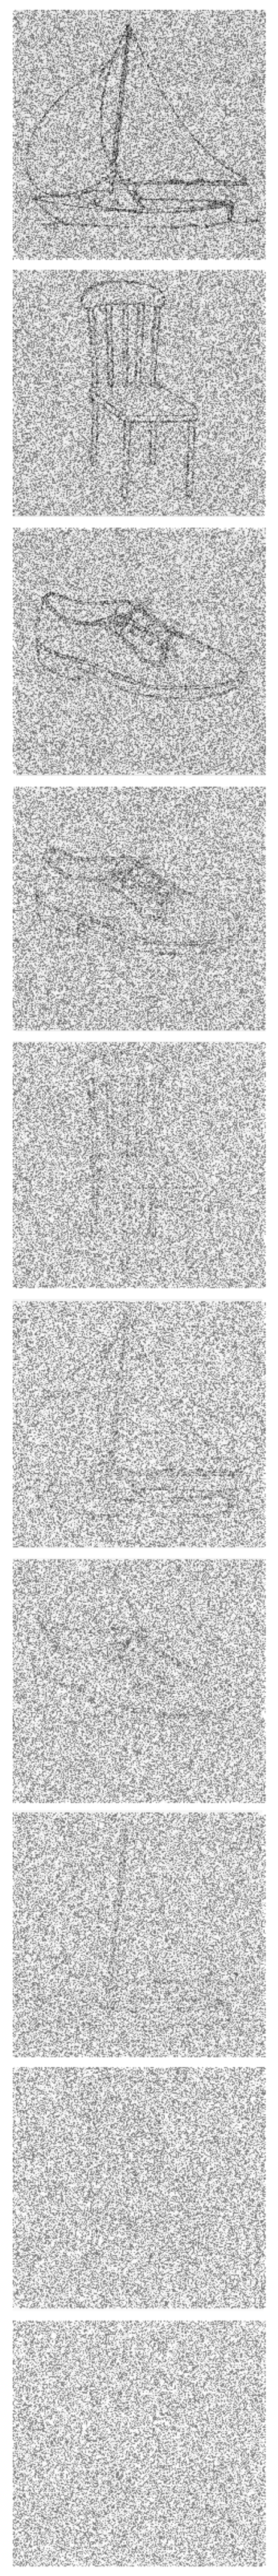

Objects

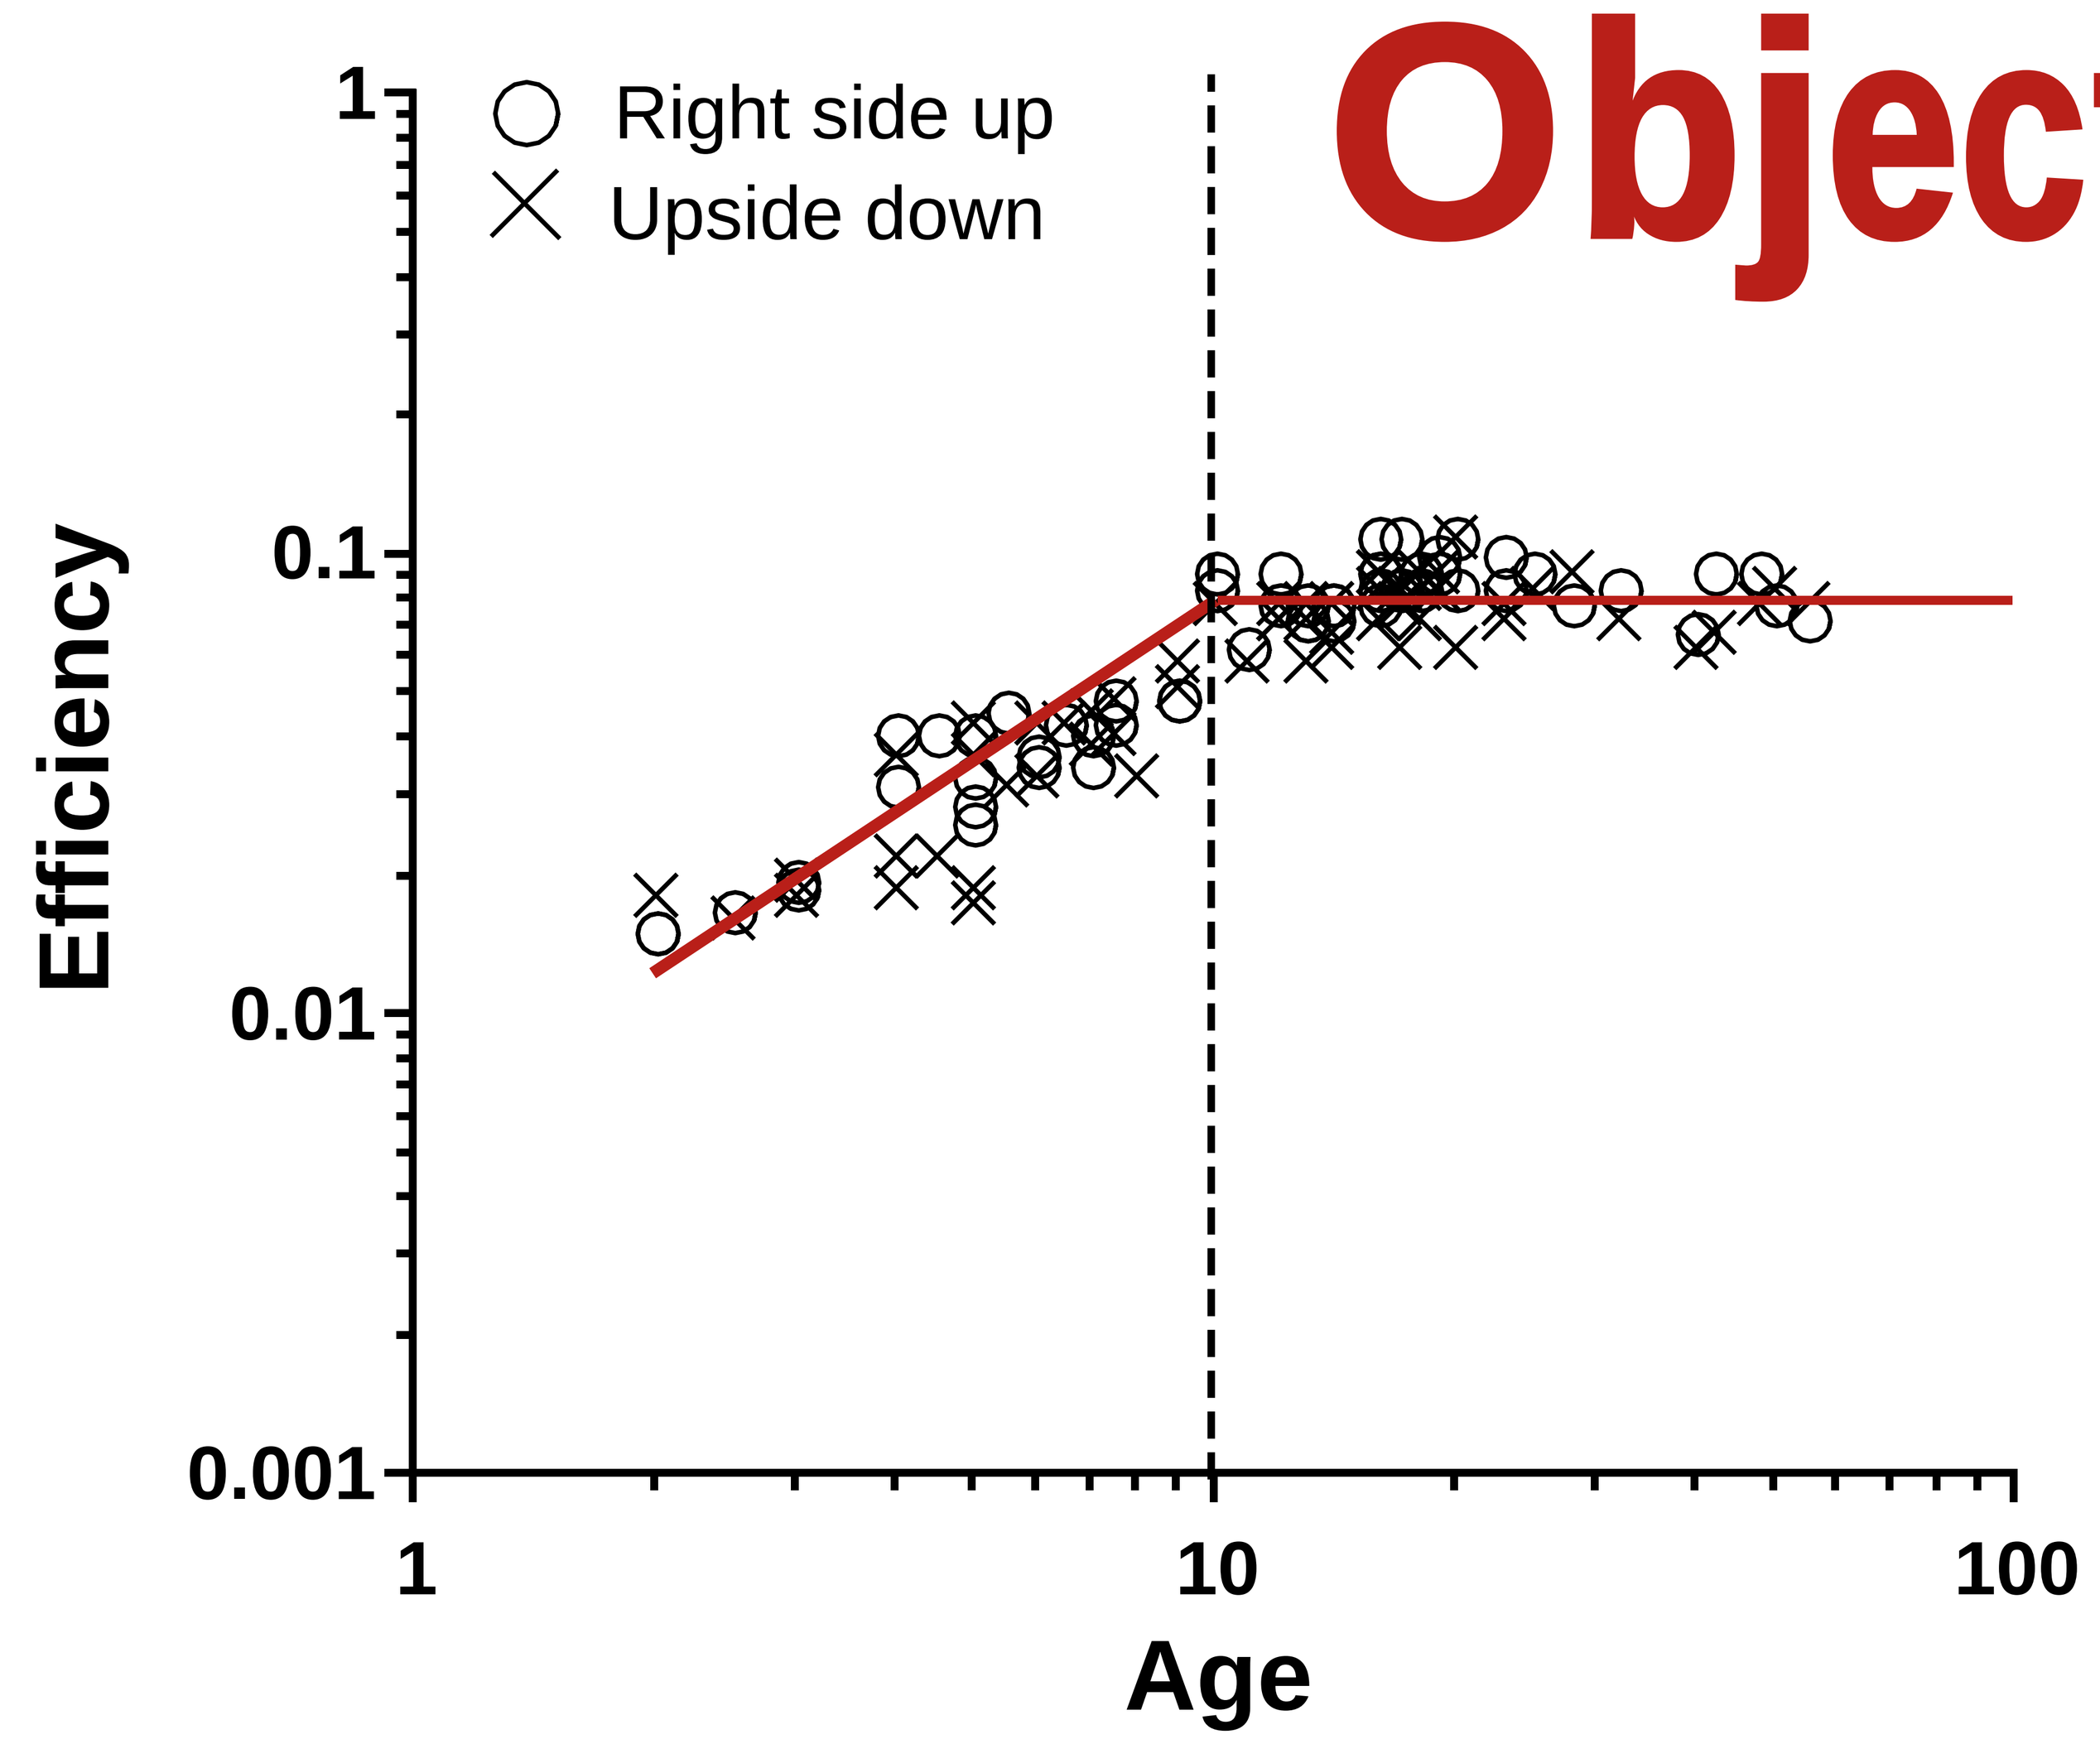
Right side up
Upside down
Efficiency
1
0.1
0.01
0.001
1
10
100
Age

METHODS: We measured efficiency for identifying letters (K, V, and D), line drawings of objects (boat, chair, and shoe), and facial expressions (happy, sad, and fearful), right-side-up and up-side-down, as a function of age (2 to 70 years old). We present the target signal on a noise background. Observers are given a reference drawing of the 3 possible targets at the same orientation, and are asked to identify each noisy stimulus by pointing at the corresponding reference image. We tested at 23 contrast levels.

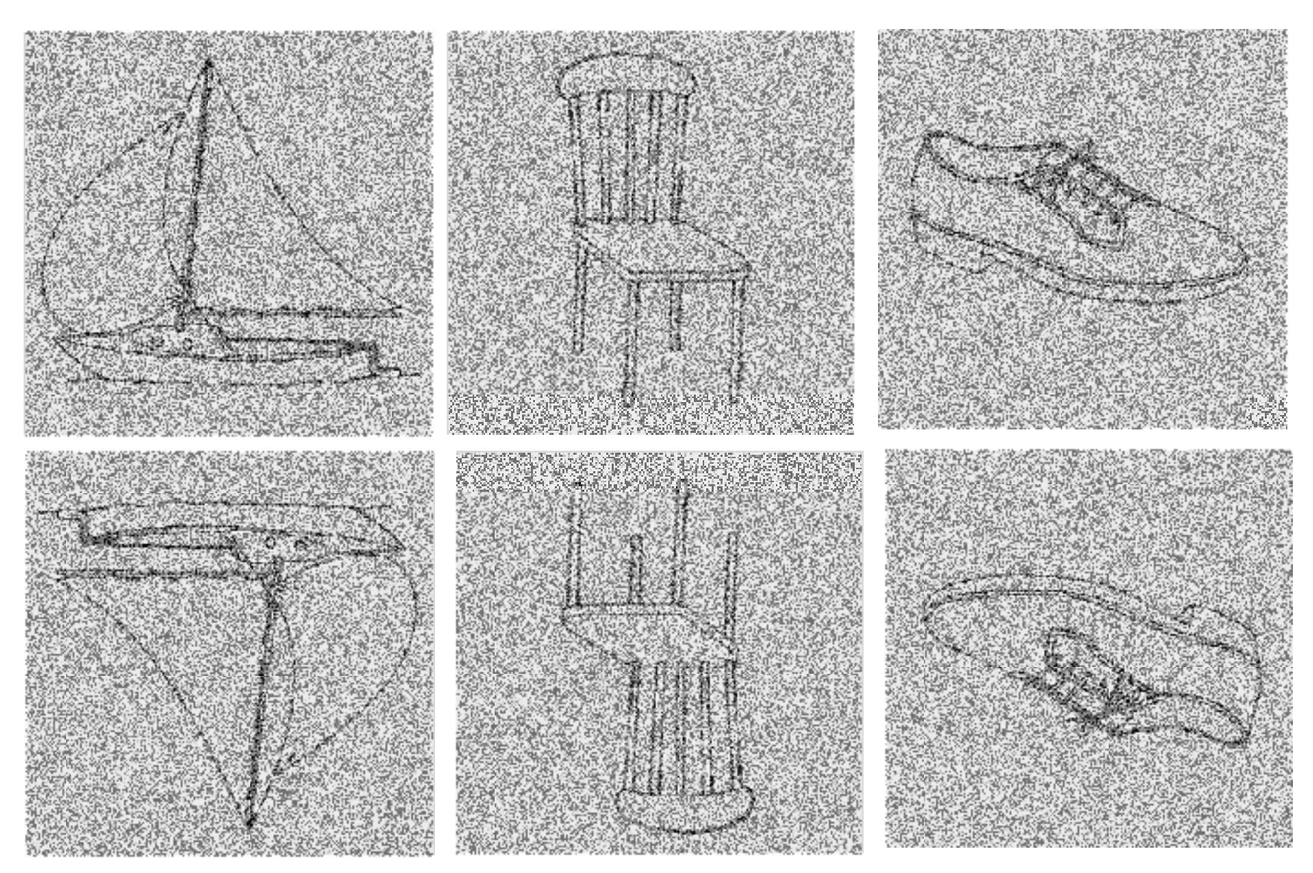

The vertical eye chart to the left uses these objects to measure your threshold.

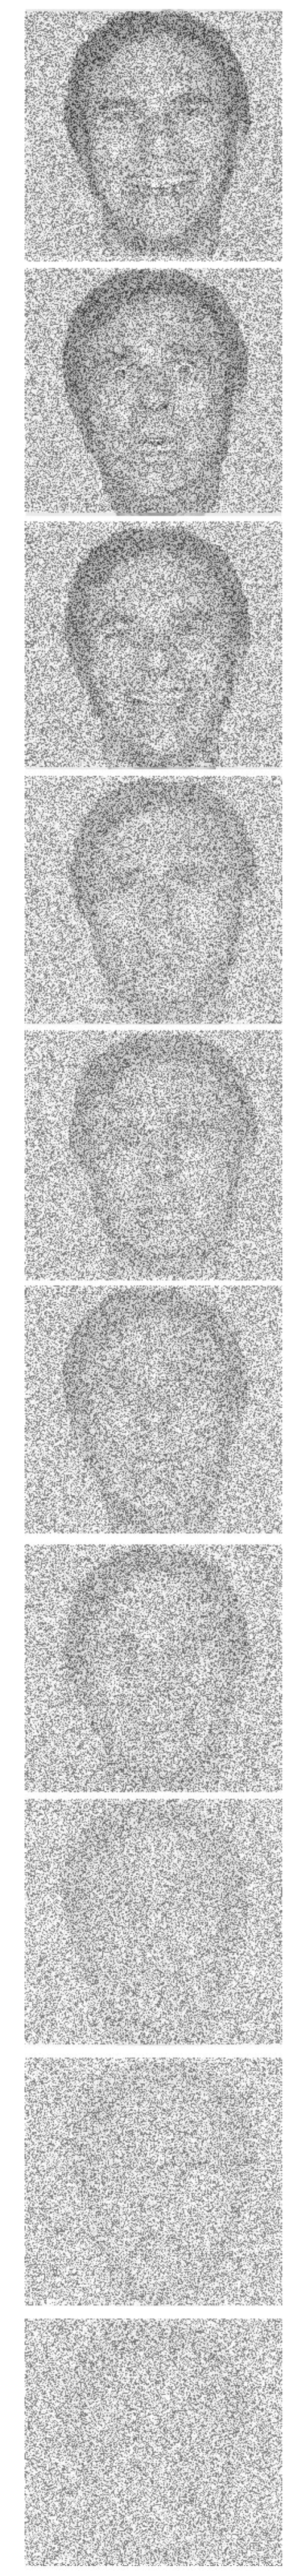

Faces

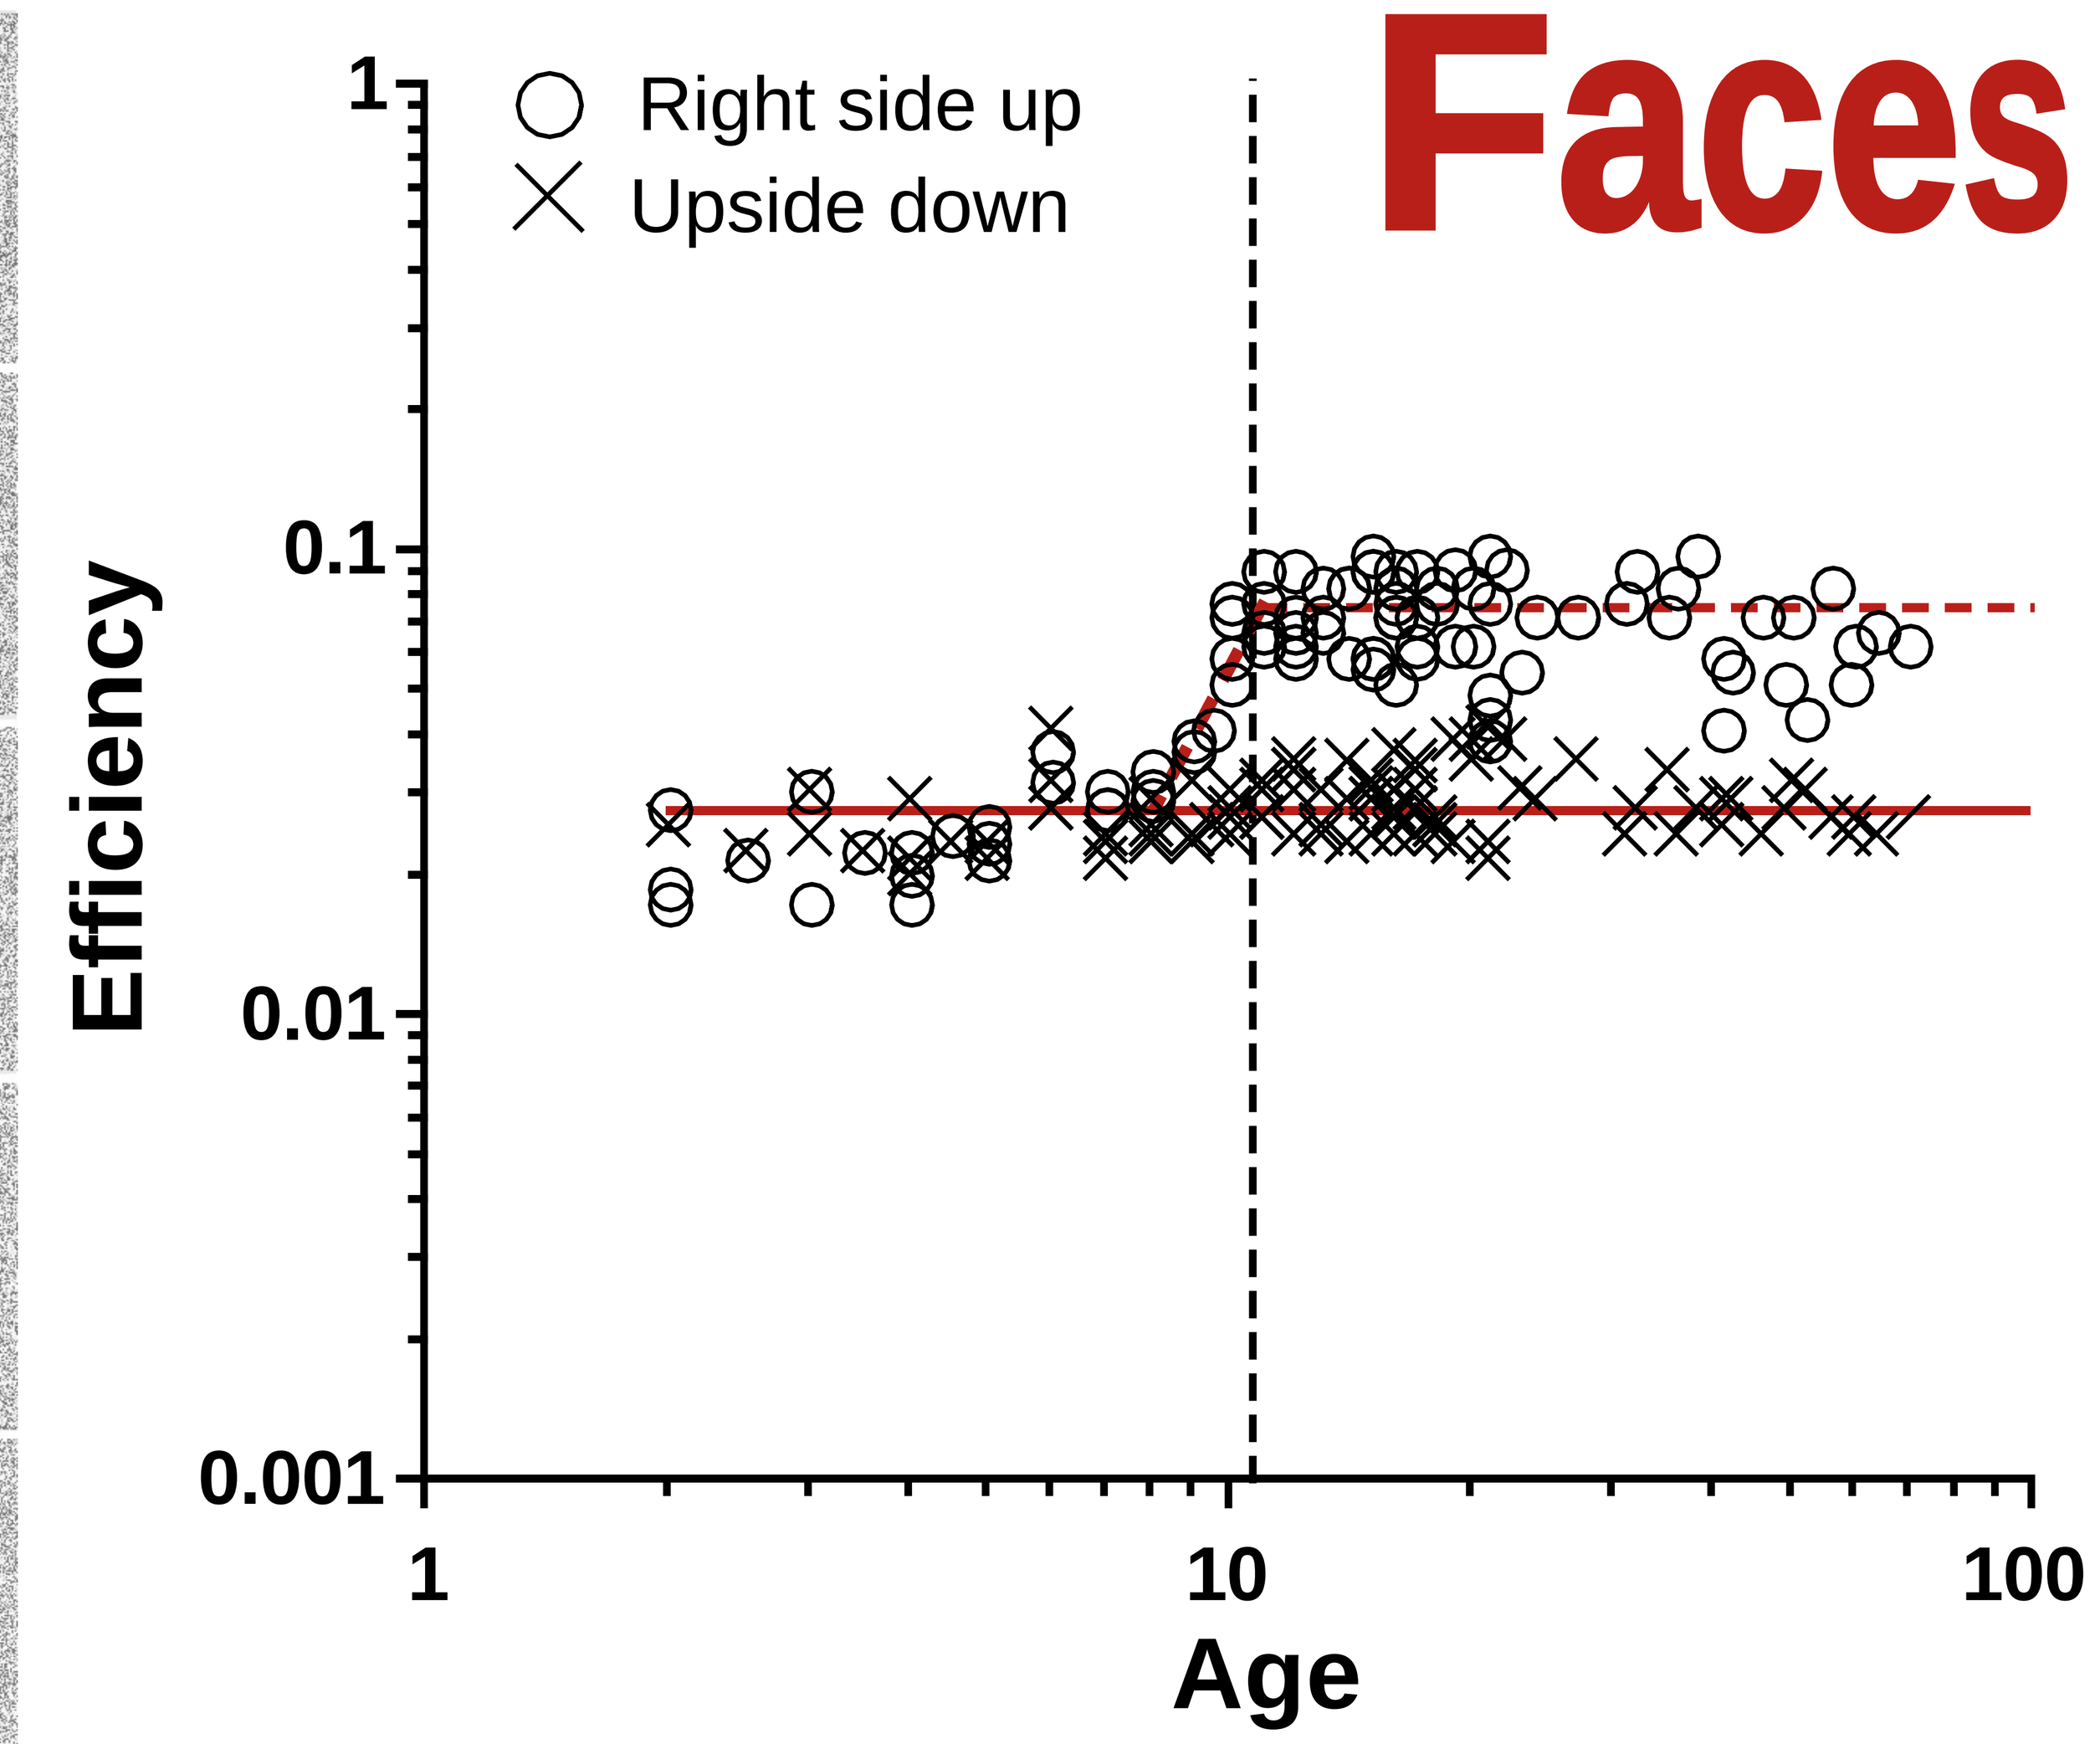
Right side up
Upside down
Efficiency
1
0.1
0.01
0.001
1
10
100
Age

RESULTS AND CONCLUSIONS: For the three kinds of target, observers reach adult efficiency at age ten, and adult efficiencies are similar (7% to 15%). However face, letter, and object recognition improve by different factors. Efficiency for identifying a letter improves 75-fold (from 0.2% to 15%). Efficiency for object recognition improves 5-fold (from 1.5% to 8%). Efficiency for identifying a facial expression improves 3-fold (from 2% to 7%) in the right-side up orientation, and doesn't improve at all in the upside-down orientation. Here, efficiency shows that faces are special.

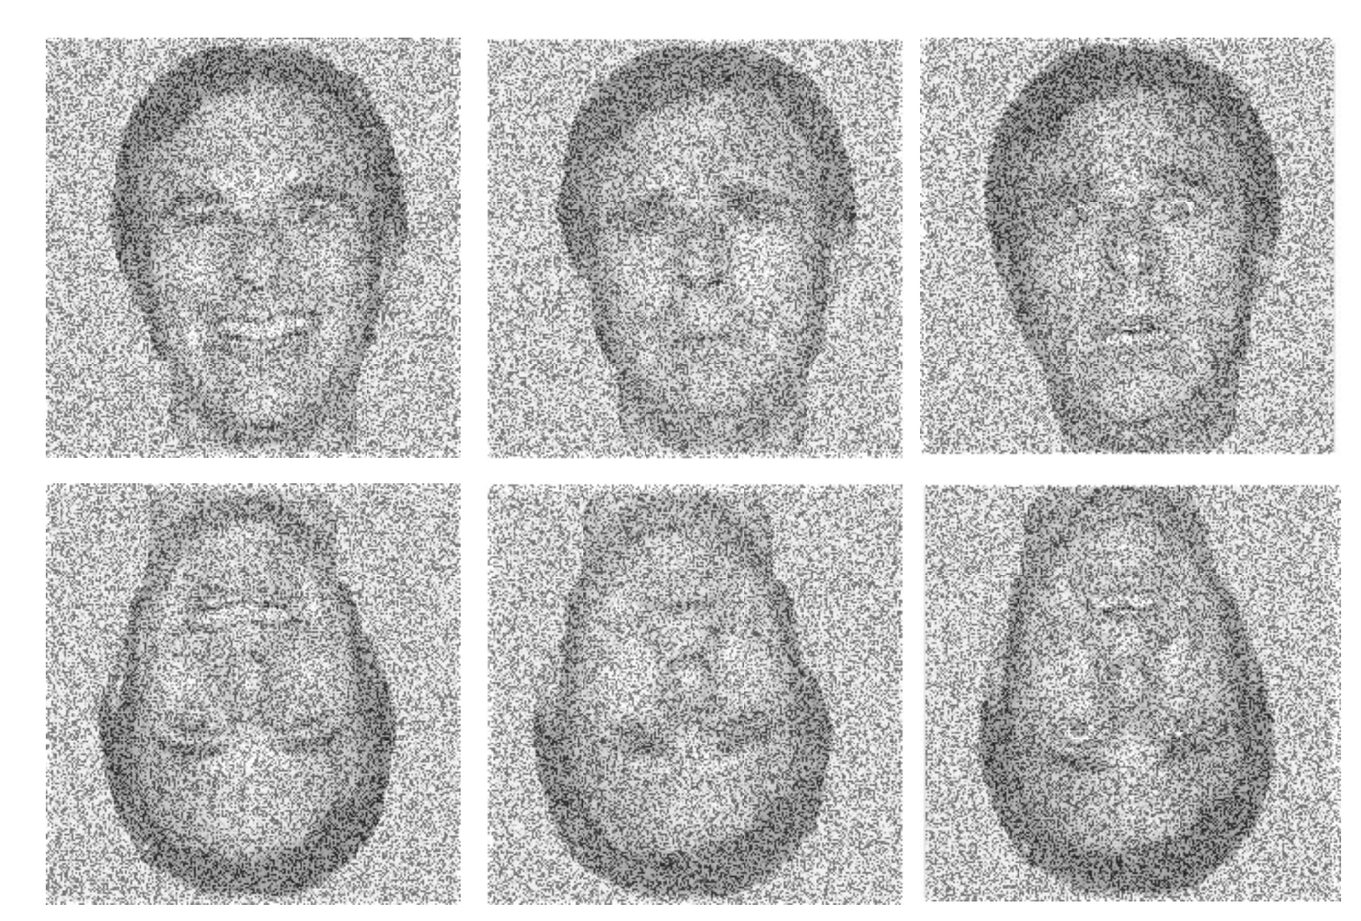

The vertical eye chart to the left uses these faces to measure your threshold.

Mishra, A., Baweja, G., Martelli, M., Chen, I., Fox, J., Majaj, N., Pelli, D.G. (2002) How efficiency for identifying objects improves with age, European Conference on Visual Perception, Glasgow, Scotland, August 25-29.
http://psych.nyu.edu/pelli/#posters