Beauty requires thought

The experience of beauty is selectively impaired by a cognitive task

NYU DAAD Aenne A. Brielmann & Denis G. Pelli

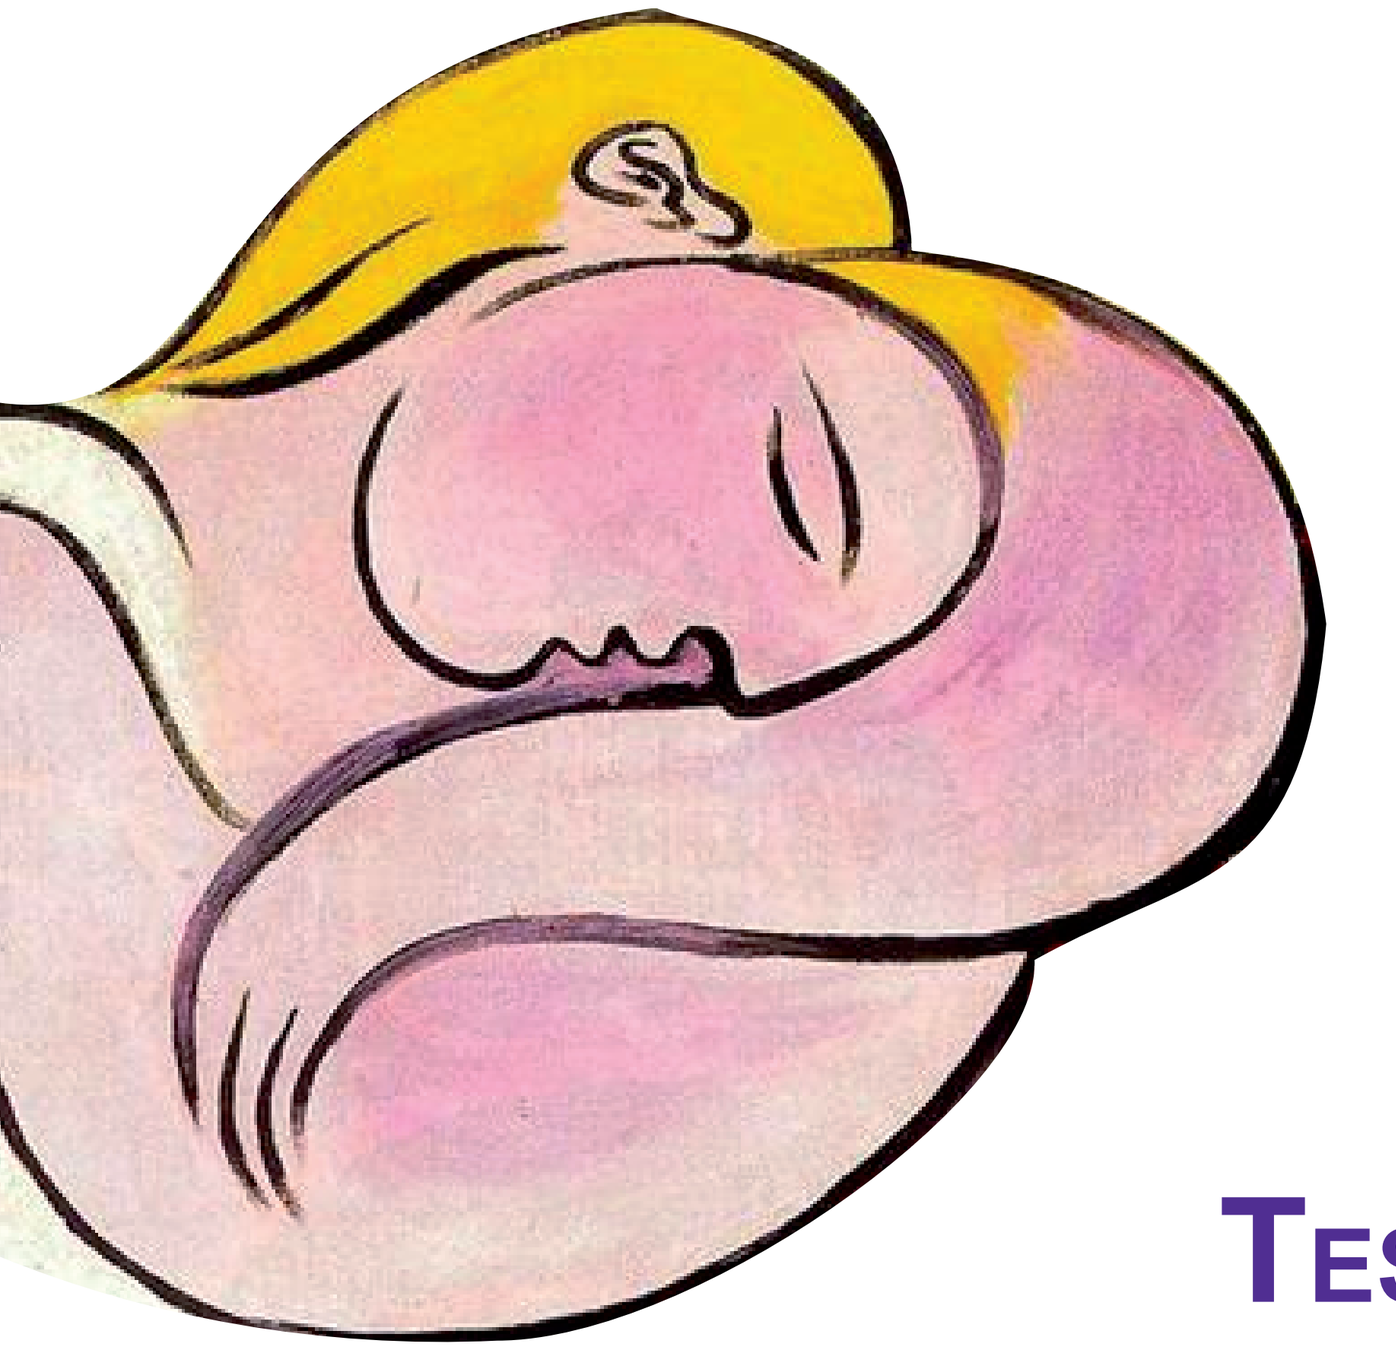

Testing Kant

The experience of beauty is a special sort of pleasure [1,2,3]

Kant: "Beauty requires thought, and ordinary pleasures do not."

Test of Kant's hypothesis:
Can beauty be experienced without cognitive resources?
Is this requirement unique to the pleasure of beauty?

Continuous pleasure ratings

Custom touchscreen application emotiontracker.com

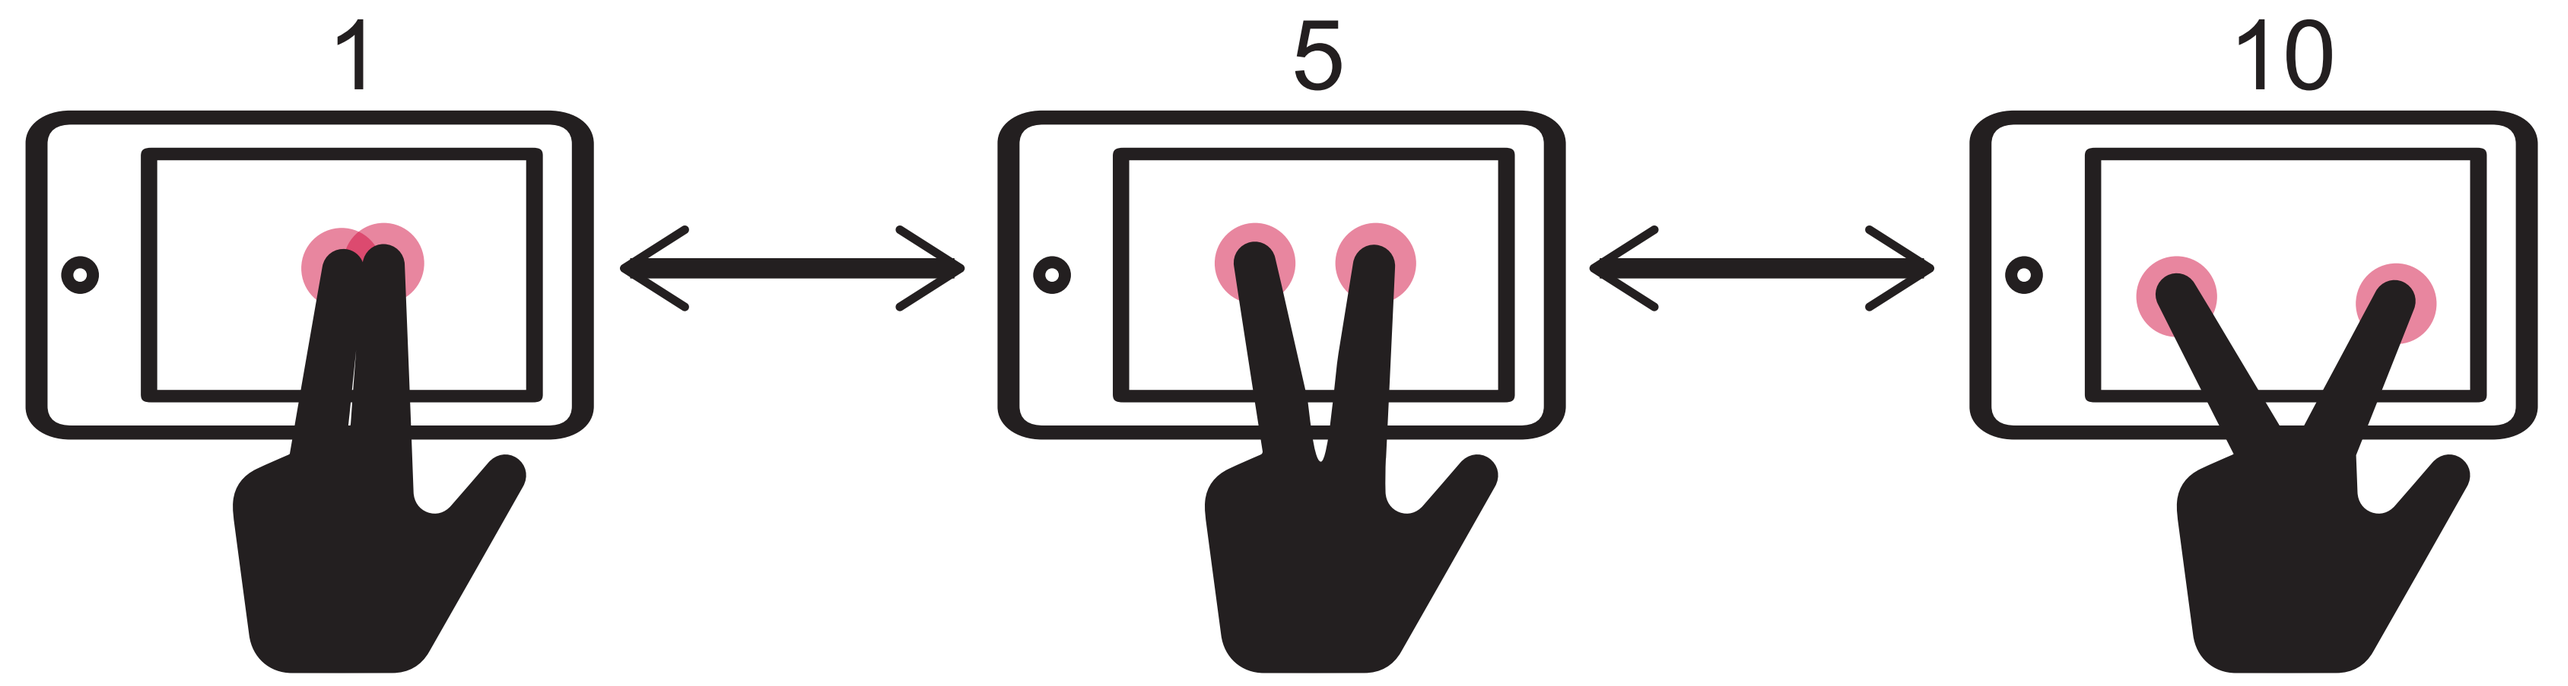

1
5
10

Trial sequence

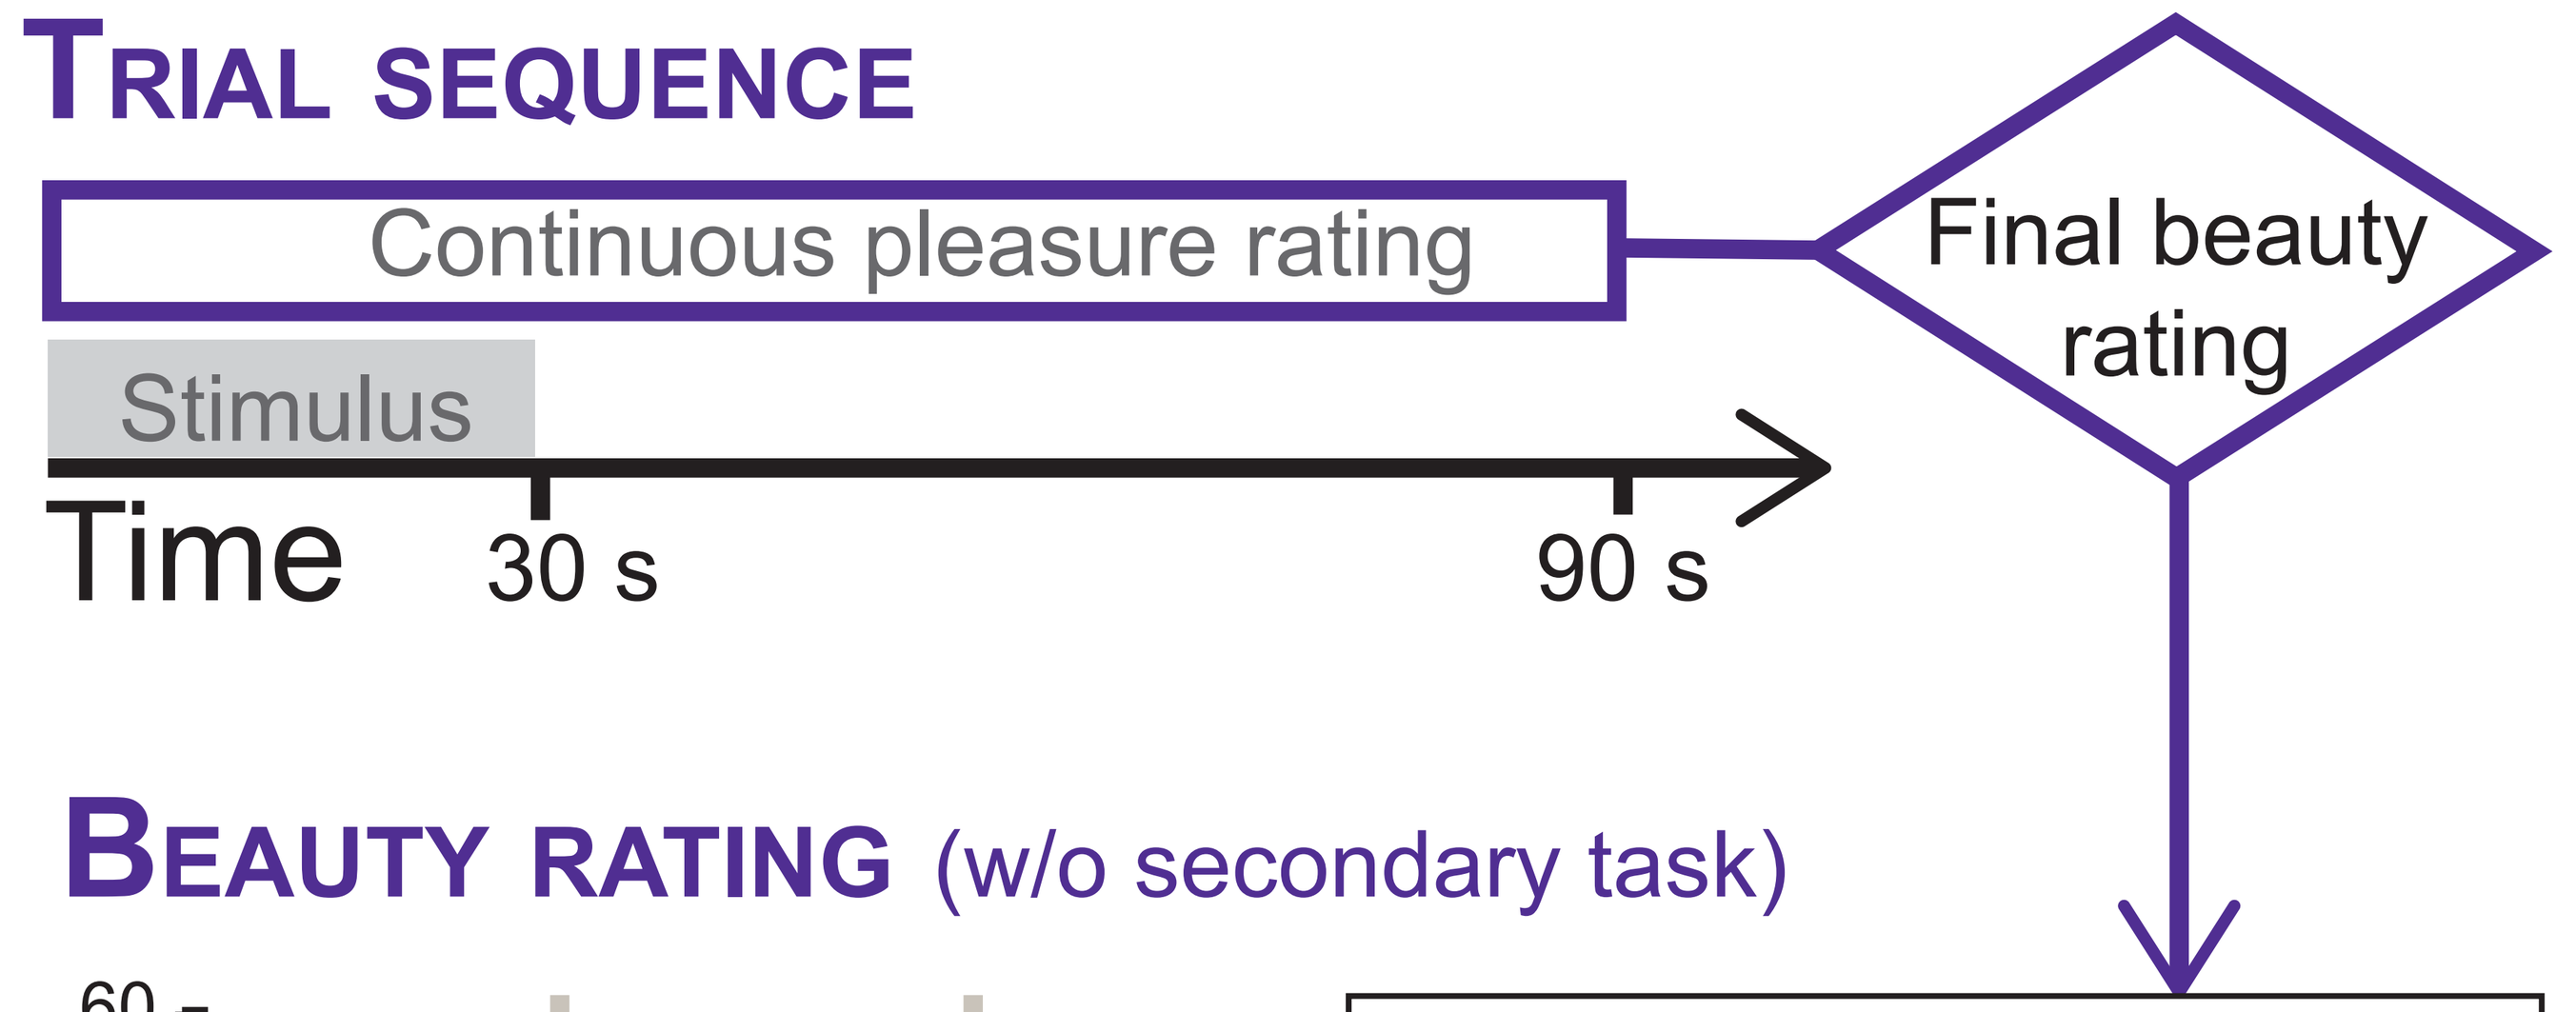

Continuous pleasure rating
Final beauty rating
Stimulus
Time
30 s
90 s

Beauty rating (w/o secondary task)

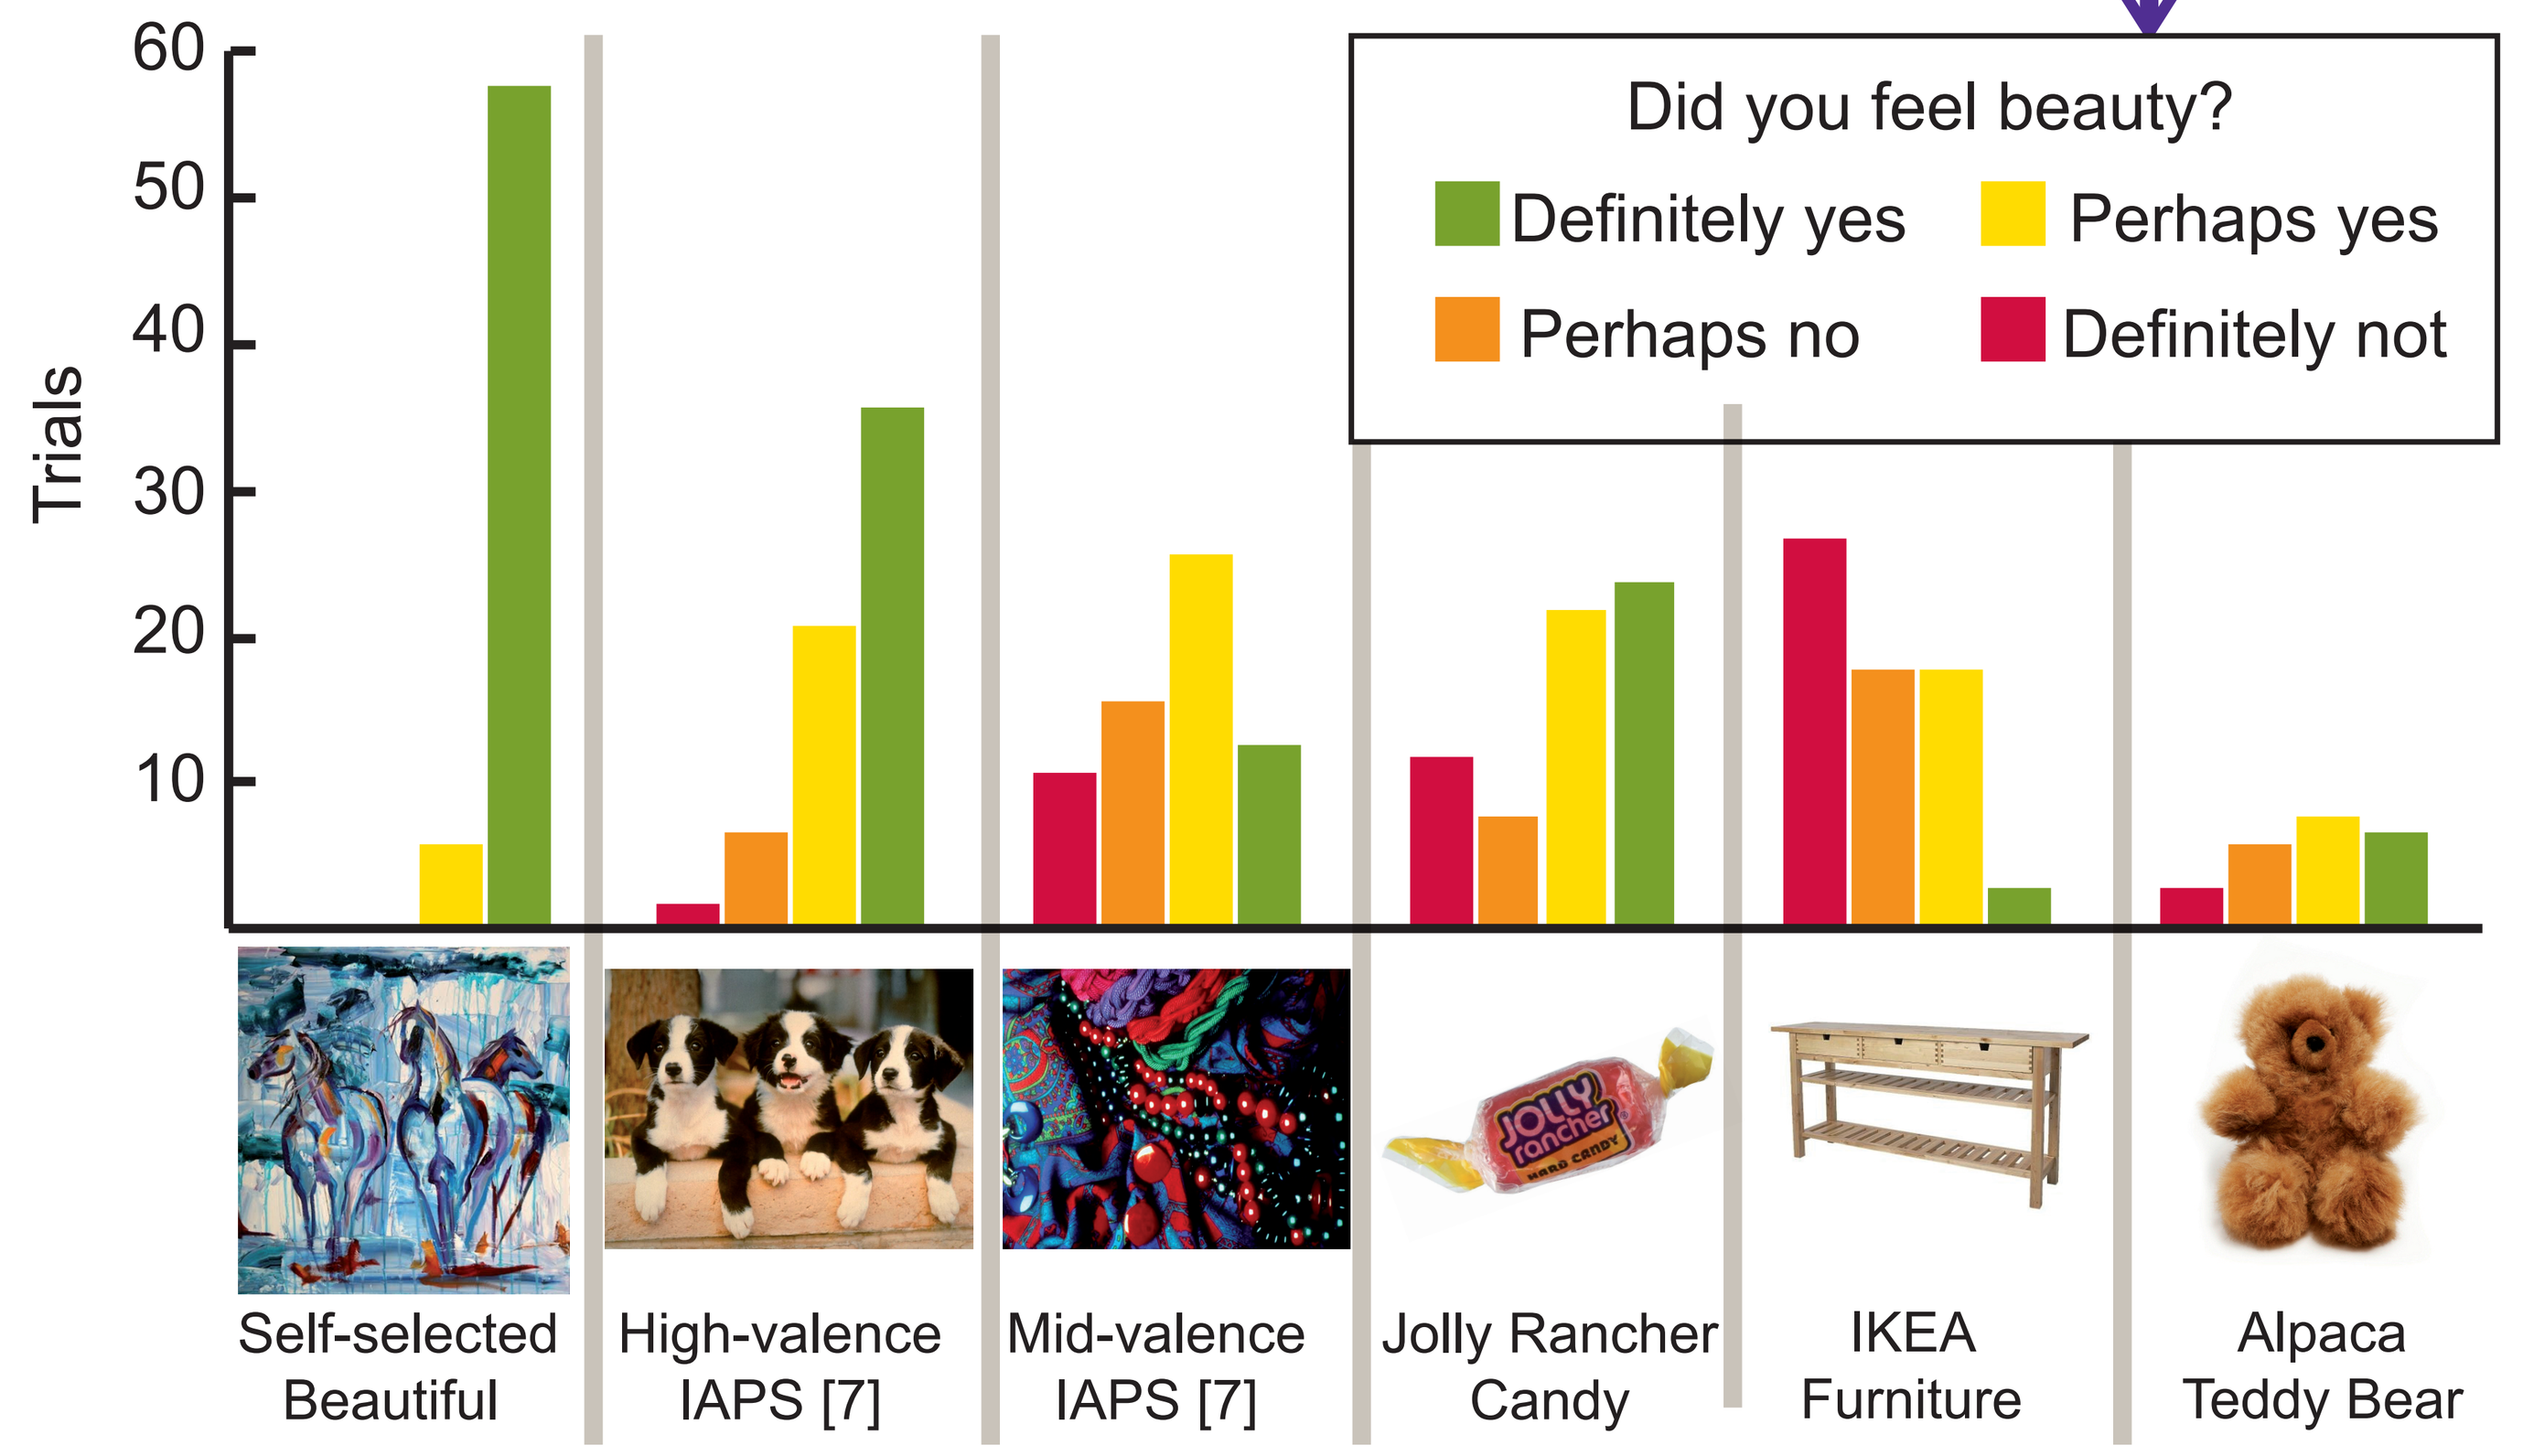

Did you feel beauty?
Definitely yes
Perhaps yes
Perhaps no
Definitely not
Trials
60
50
40
30
20
10
Self-selected Beautiful
High-valence IAPS [7]
Mid-valence IAPS [7]
Jolly Rancher Candy
IKEA Furniture
Alpaca Teddy Bear

Only pleasure from beautiful stimuli is reduced while performing a secondary cognitive task.

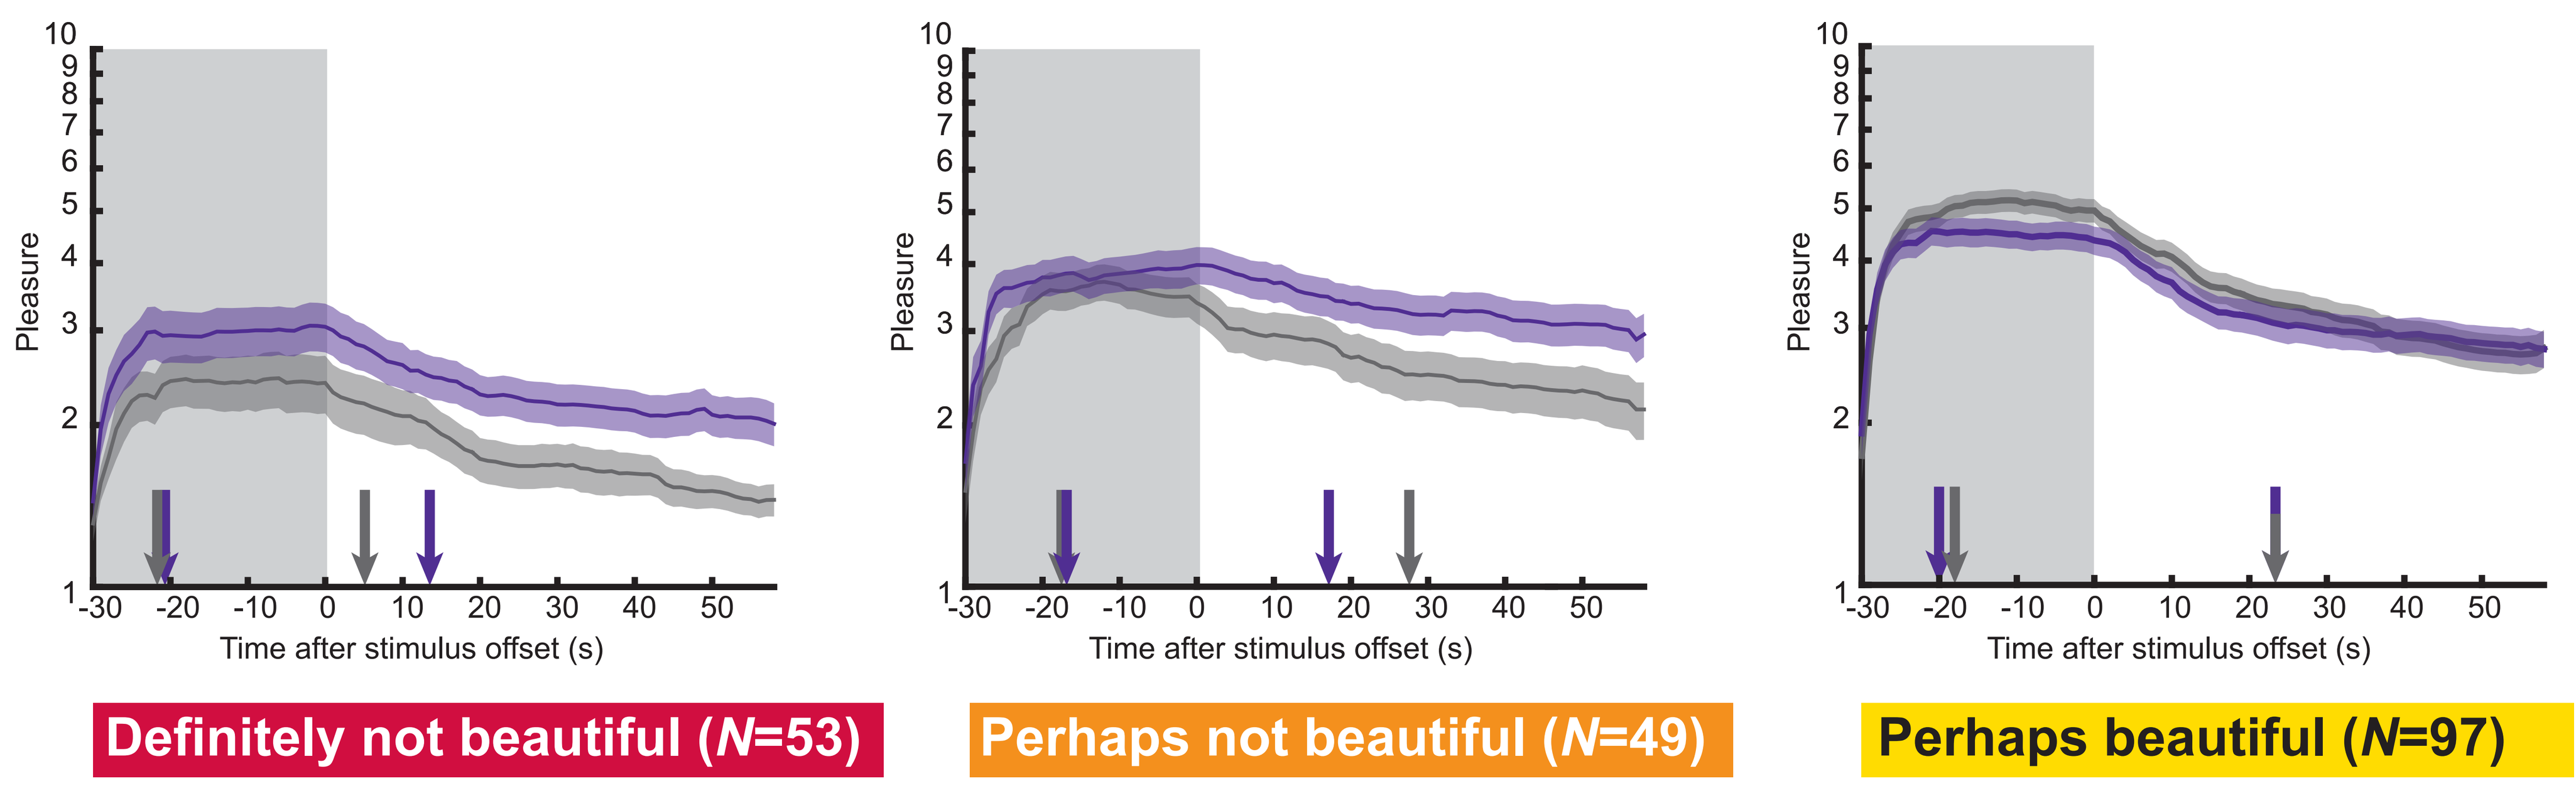

Pleasure
Time after stimulus offset (s)
-30 -20 -10 0 10 20 30 40 50
Definitely not beautiful (N=53)
Perhaps not beautiful (N=49)
Perhaps beautiful (N=97)

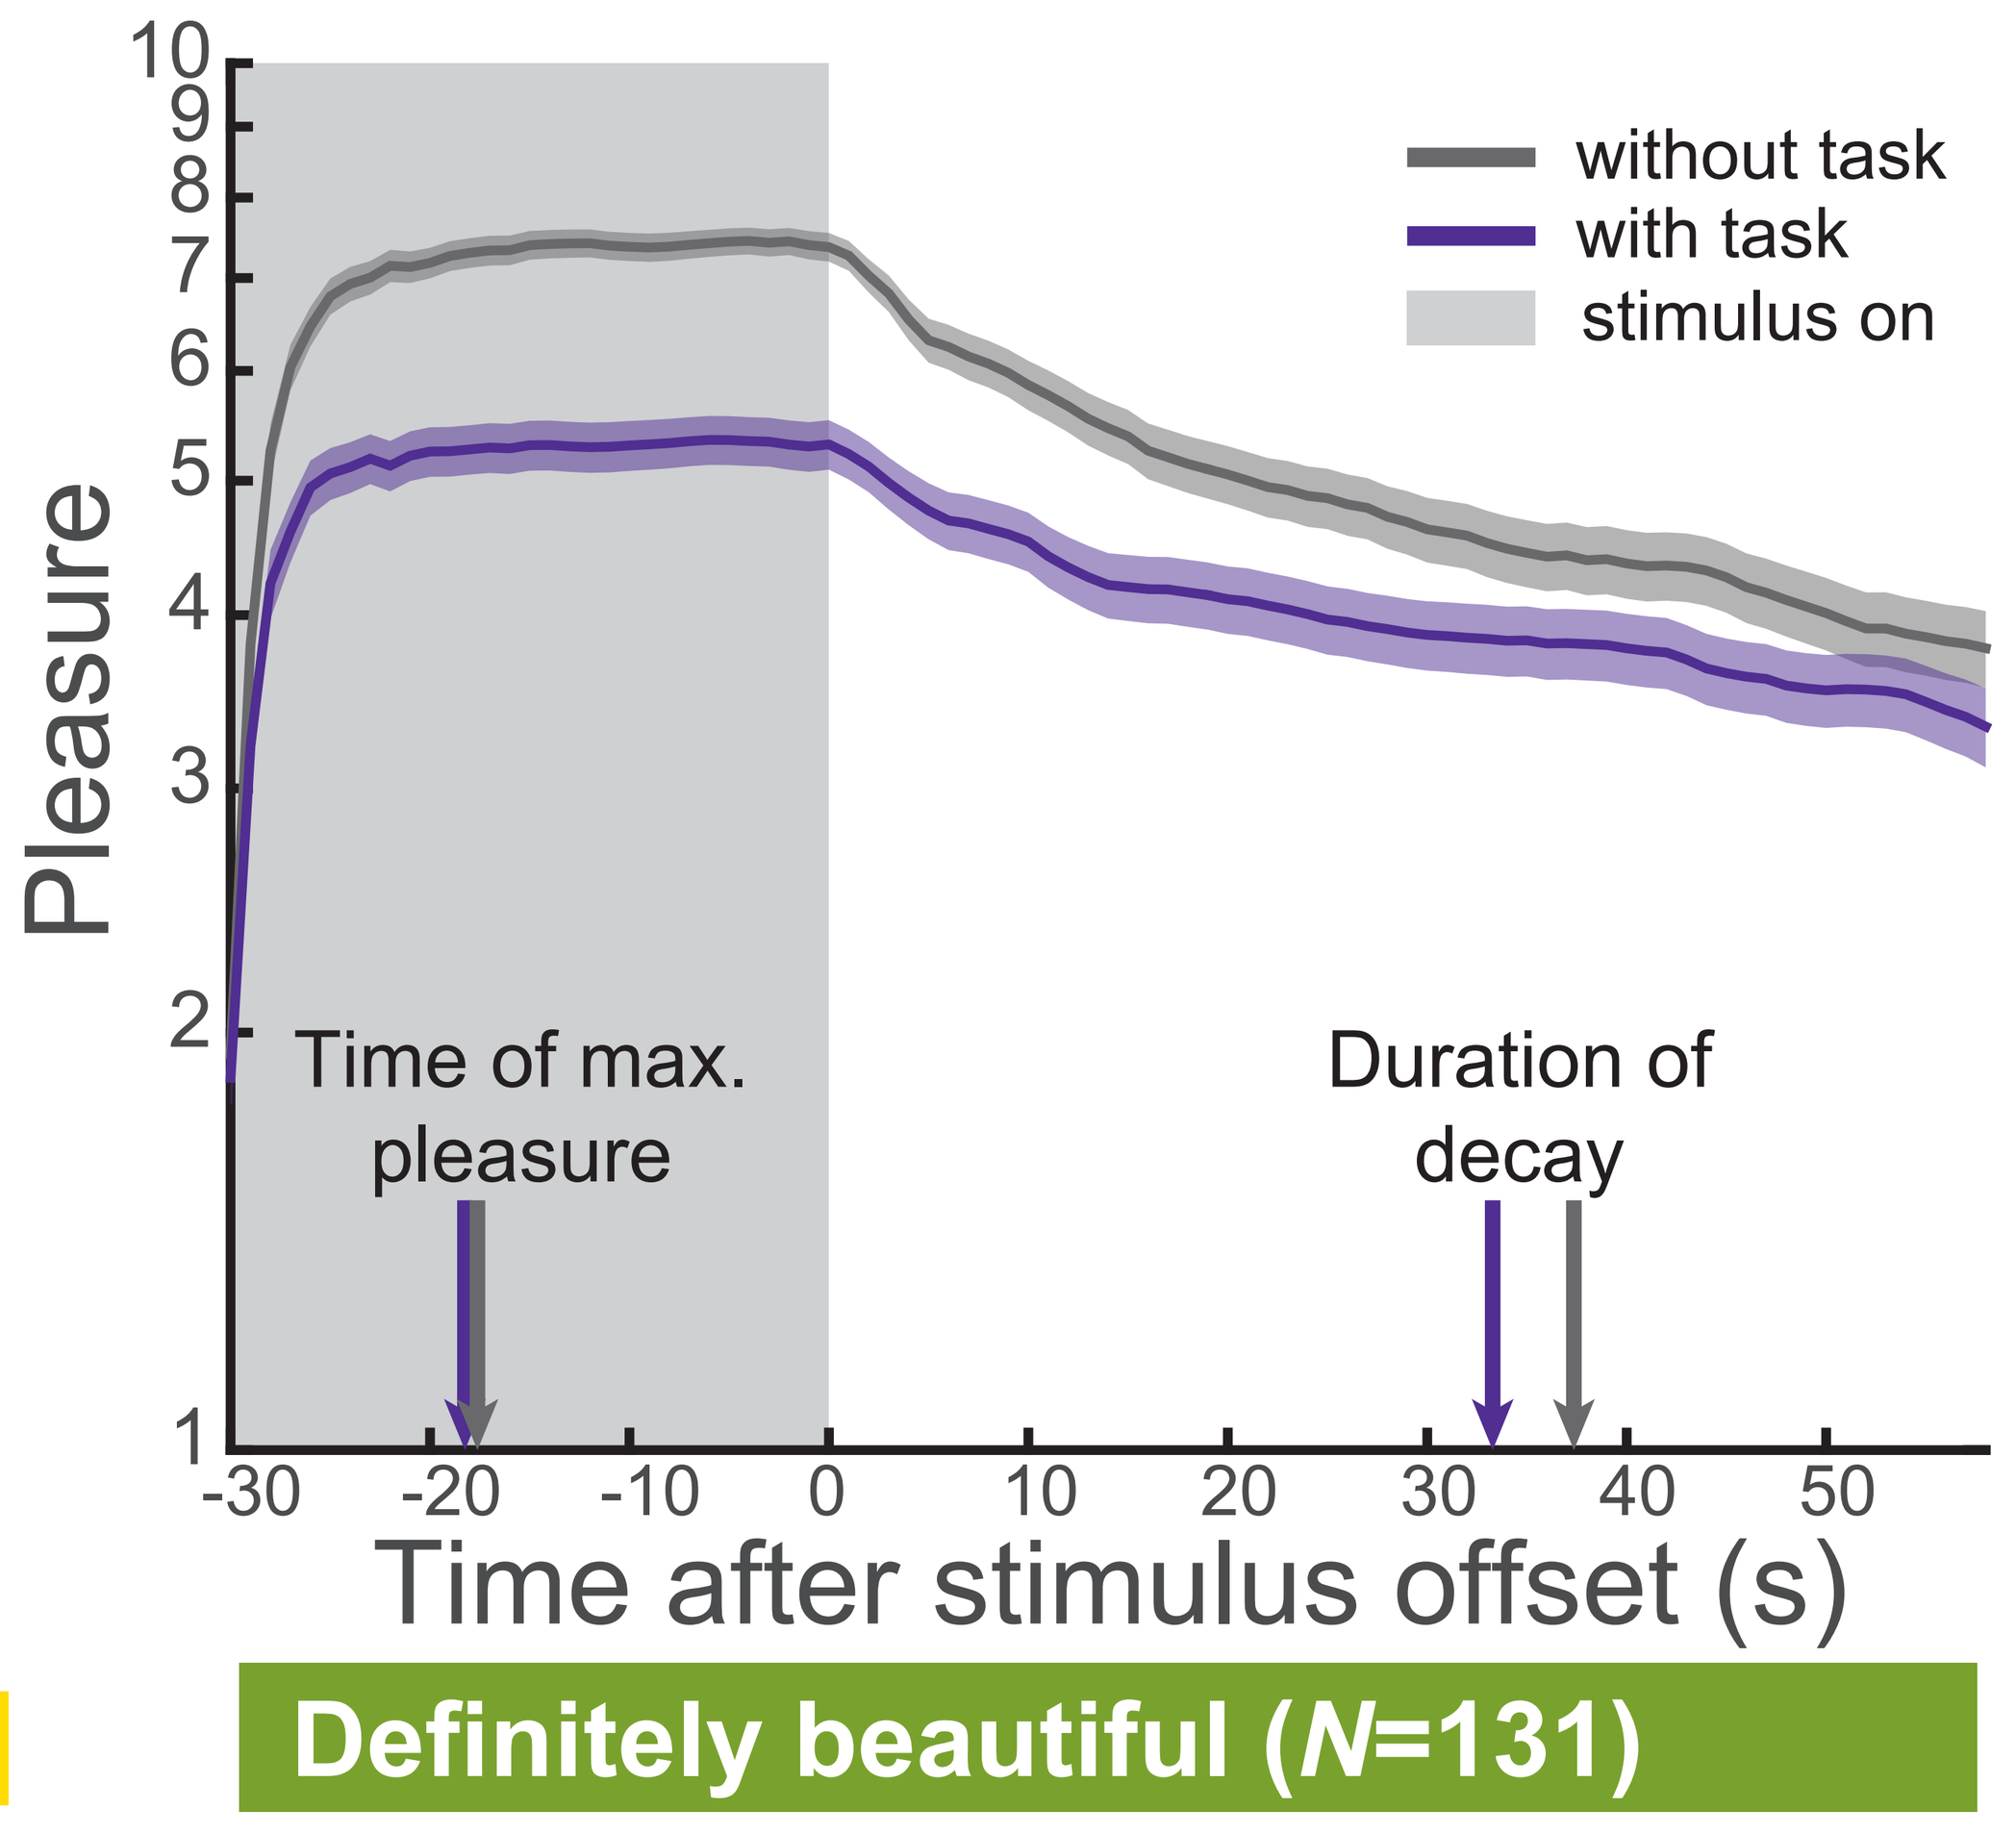

without task
with task
stimulus on
Pleasure
Time of max. pleasure
Duration of decay
Time after stimulus offset (s)
-30 -20 -10 0 10 20 30 40 50
Definitely beautiful (N=131)

Experiment 1

Experiment 2

2-back task (letters)

L S R S T T

Key press response for each target letter (Experiment 1)

Verbal response for each target letter (Experiment 2)

Experiment 3

Digit span task

4 5 0 2 8 5 3 3 7 8

Verbal response once at the end of each trial

Cognitive load reduces pleasure of beautiful but not ordinary experiences, both during ...

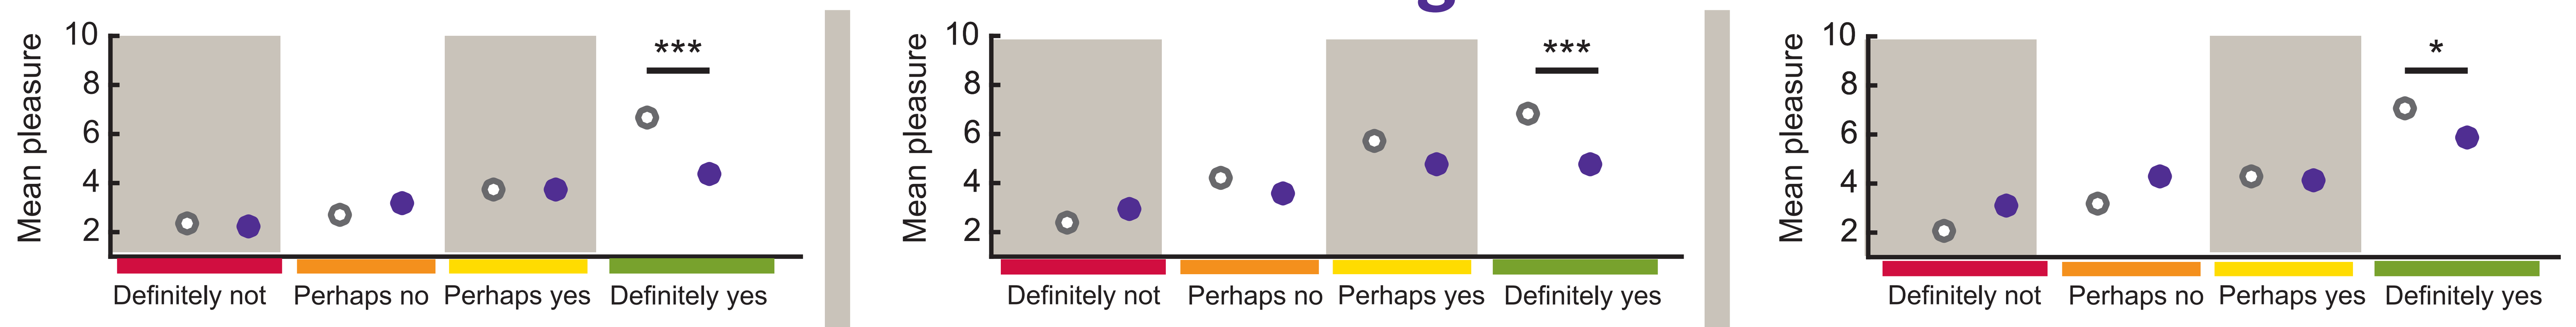

Mean pleasure
10
8
6
4
2

*
Definitely not
Perhaps no
Perhaps yes
Definitely yes
w/o task
with task

and after stimulus.

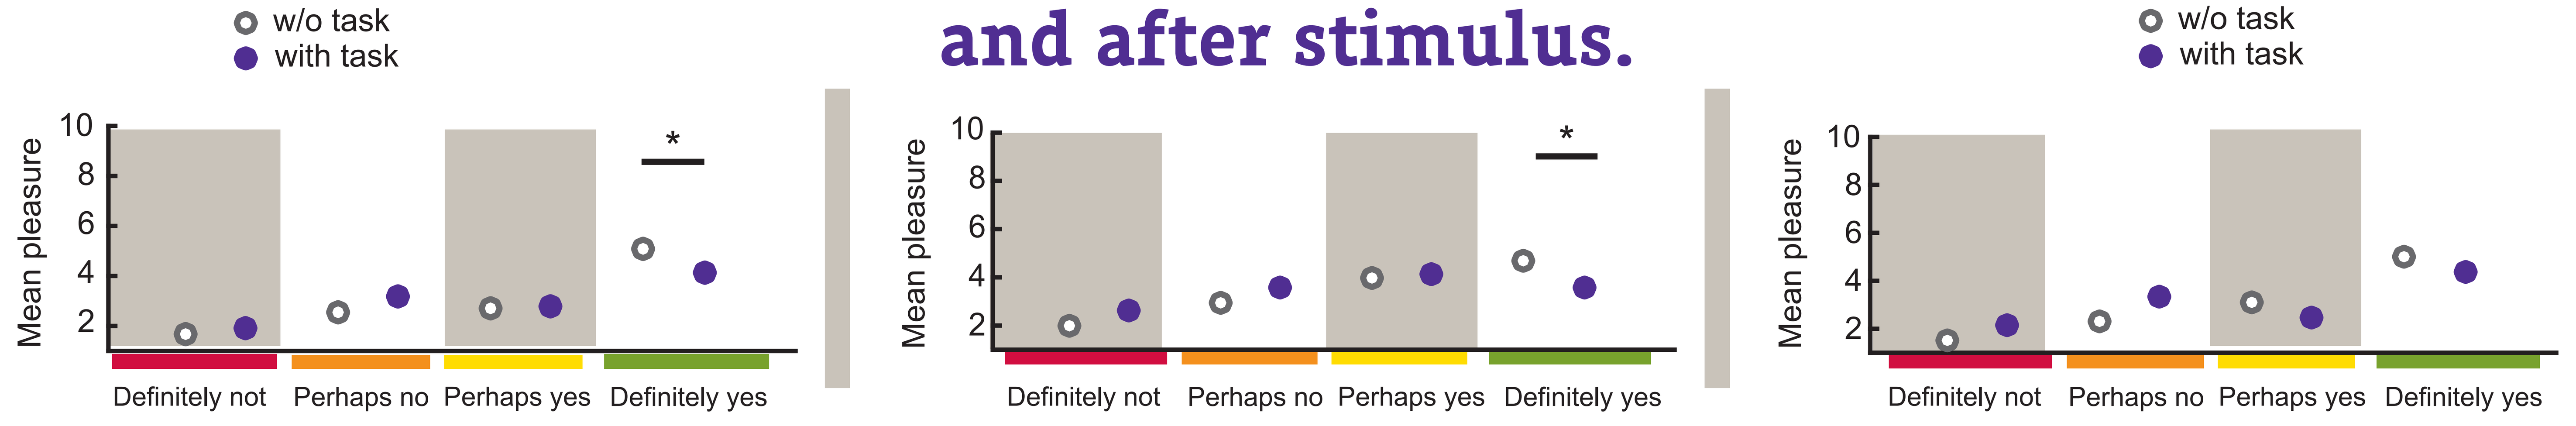

Mean pleasure
10
8
6
4
2
*
*
Definitely not
Perhaps no
Perhaps yes
Definitely yes
w/o task
with task

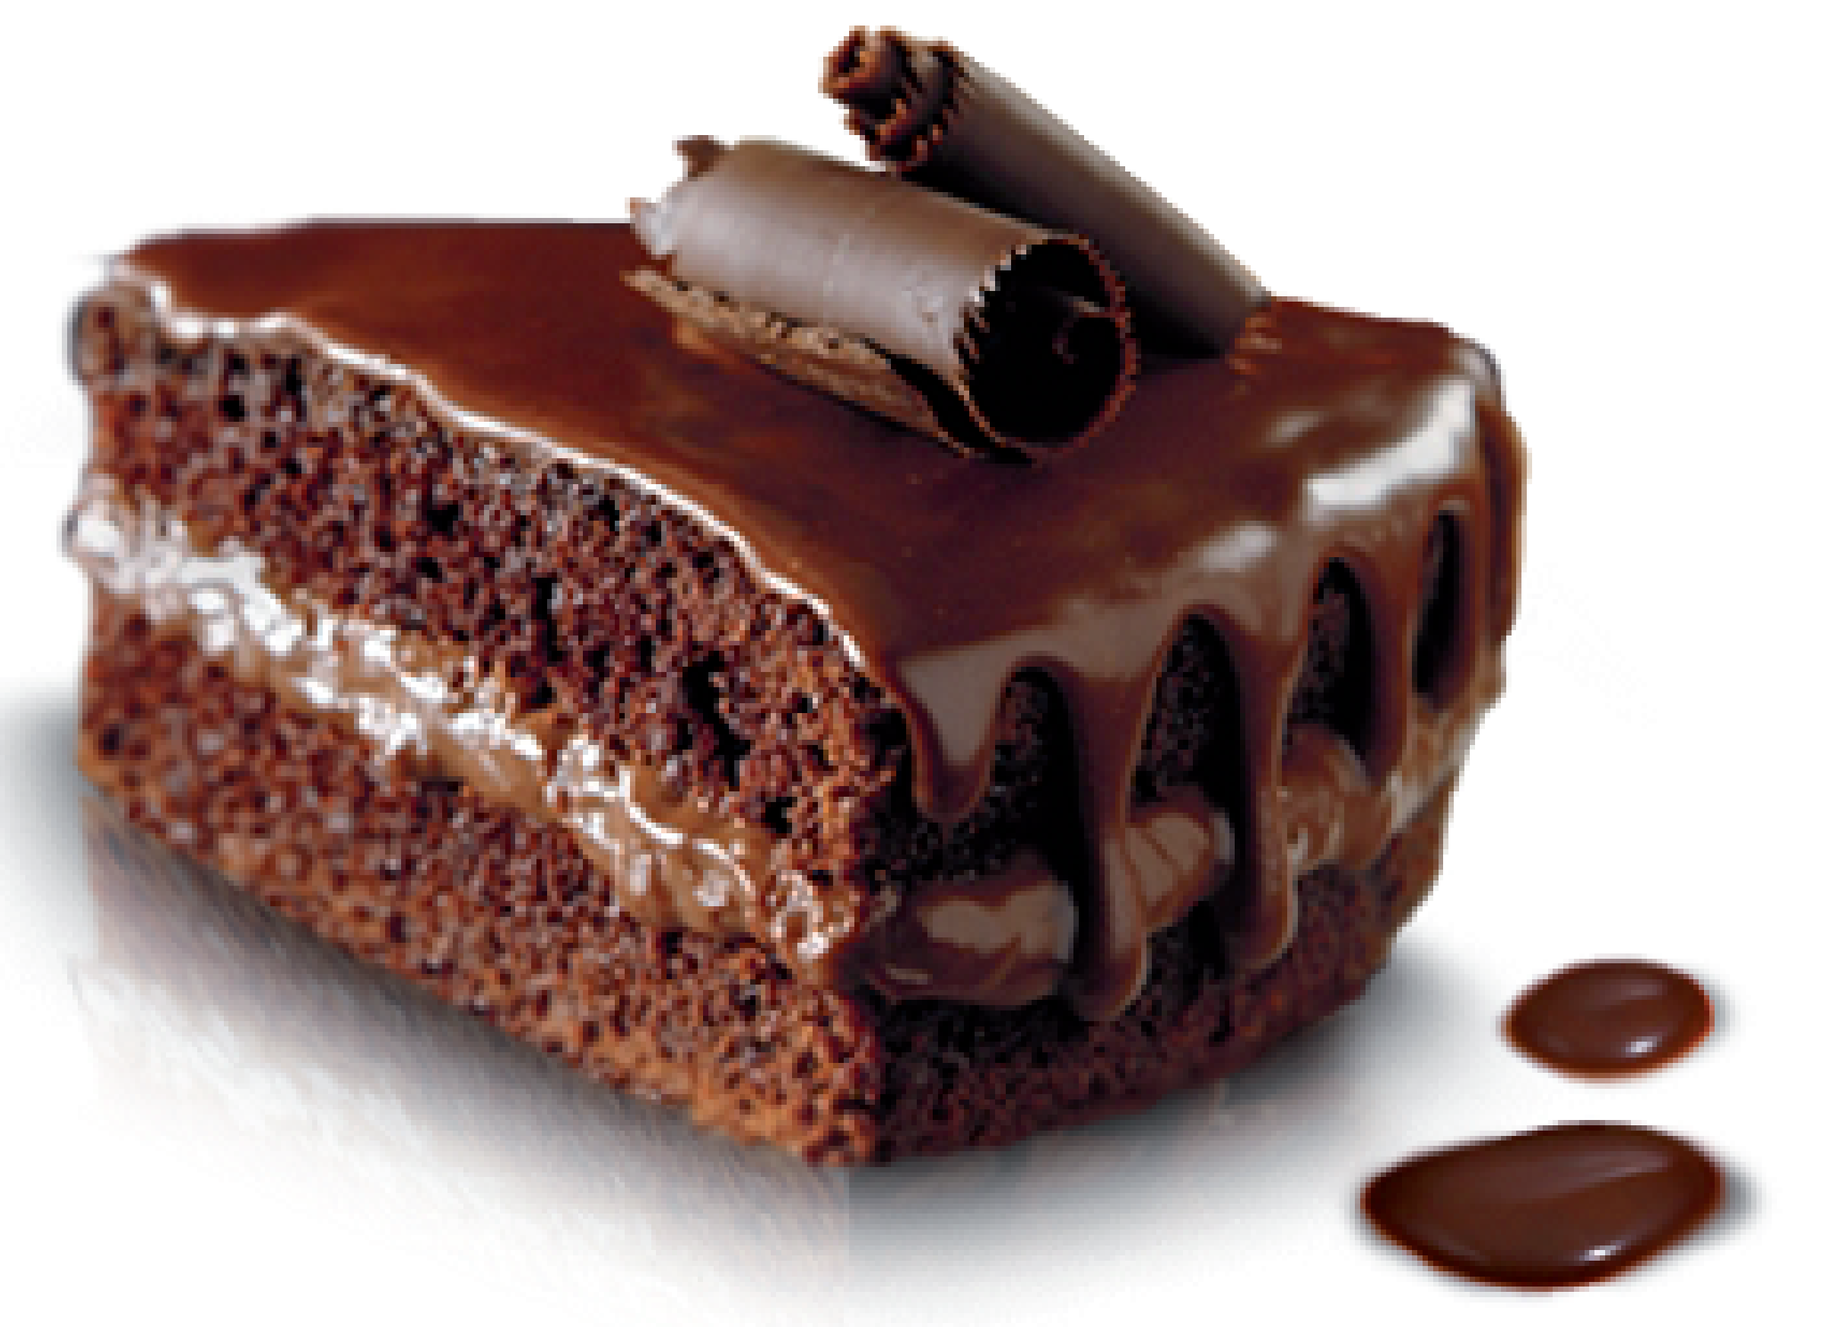

Discussion

The experience of beauty is characterized by high pleasure ratings during and after stimulus exposure, confirming earlier findings [7].

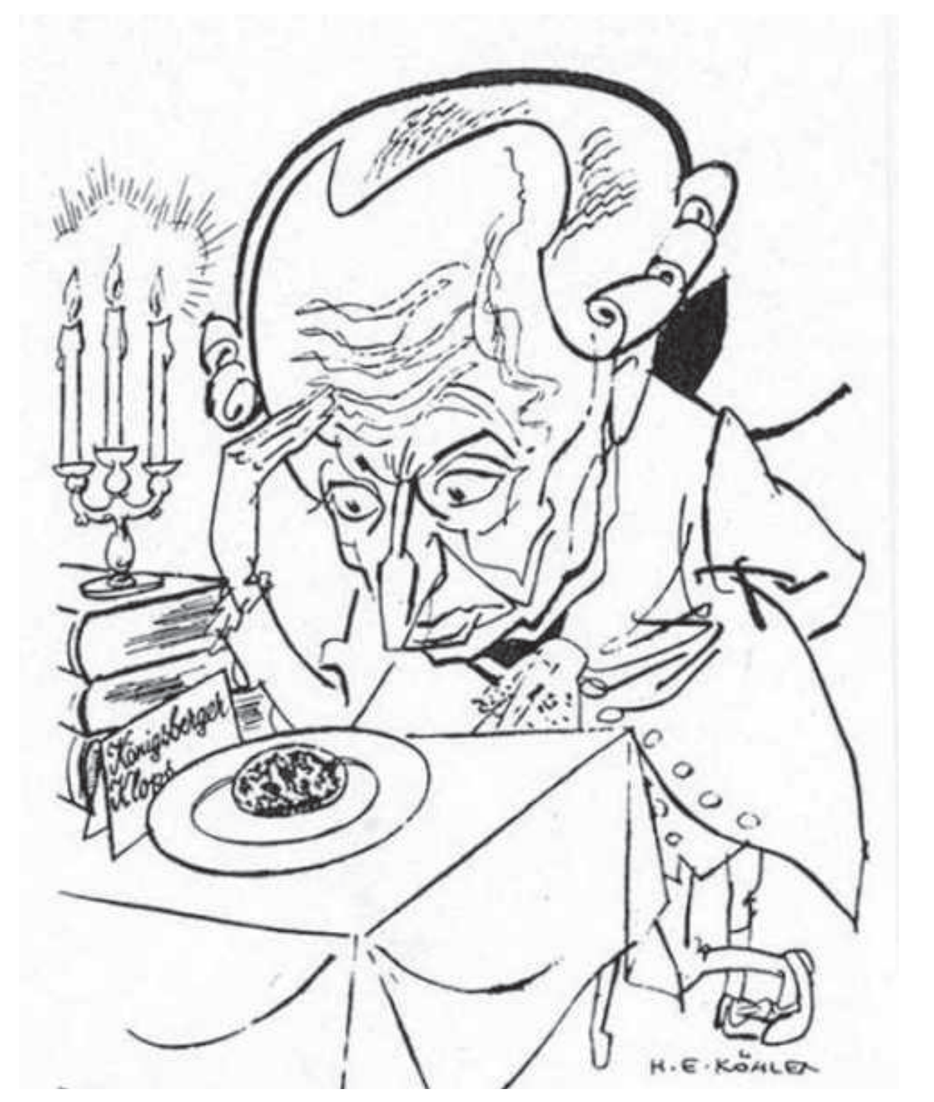

Surprisingly, people can experience beauty through touch and taste.

Kant was right: Beauty does require thought, while ordinary pleasures do not.

Future directions

Are there other obstacles to the beauty experience?
Does clinical depression block access?
What about drug addiction?

Is the integration of emotion and cognition crucial for other domains such as complex decision making?

References

1. Brown, S., Gao, X., Tisdelle, L., Eickhoff, S. B. & Liotti, M. Naturalizing aesthetics: brain areas for aesthetic appraisal across sensory modalities. *Neuroimage*, 58, 250–8 (2011).
2. Kant, I. *Kritik der Urteilskraft*. (Reclam, 1790).
3. Santayana, G. *The sense of beauty: Being the outline of aesthetic theory*. (Charles Scribner's sons, 1896).
4. Lang, P. J., Bradley, M. M. & Cuthbert, B. N. *International affective picture system (IAPS): Affective ratings of pictures and instruction manual*. (2008).
5. Vale, L., Gerger, G., Leder, H. & Pelli, D. Correlating Beauty and Two Measures of Pleasure. *J. Vis.*, 15, 246 (2015).

Acknowledgments

Funding: Deutscher Akademischer Austauschdienst (DAAD)
New York University, Graduate School of Arts and Sciences
Data collection, helpful suggestions and feedback:
Heon (Peter) Joo, Elisabeth Wente
Feedback: Lauren Vale, Amy Belfi, Apollinaire Scherr

Citation

Brielmann, A. A. & Pelli, D. G. Beauty requires thought: The experience of beauty is selectively impaired by a cognitive task. *23rd Annual Meeting of the Cognitive Neuroscience Society*, New York City, New York, April 2-5, 2016.
https://psych.nyu.edu/pelli/posters.html • https://aennebrielmann.wordpress.com/publications