THE ROLE OF EXPOSURE DURATION IN THE EXPERIENCE OF BEAUTY

Lauren Vale & Denis G. Pelli

NYU

Question

How long must one look at an image to experience beauty? In museums, the average looking time for a painting is 27 - 38 s [1,2]. Using a continuous measure of self-reported pleasure, we examined how the exposure duration of a stimulus affects the experience of beauty.

Stimuli

Beautiful personal

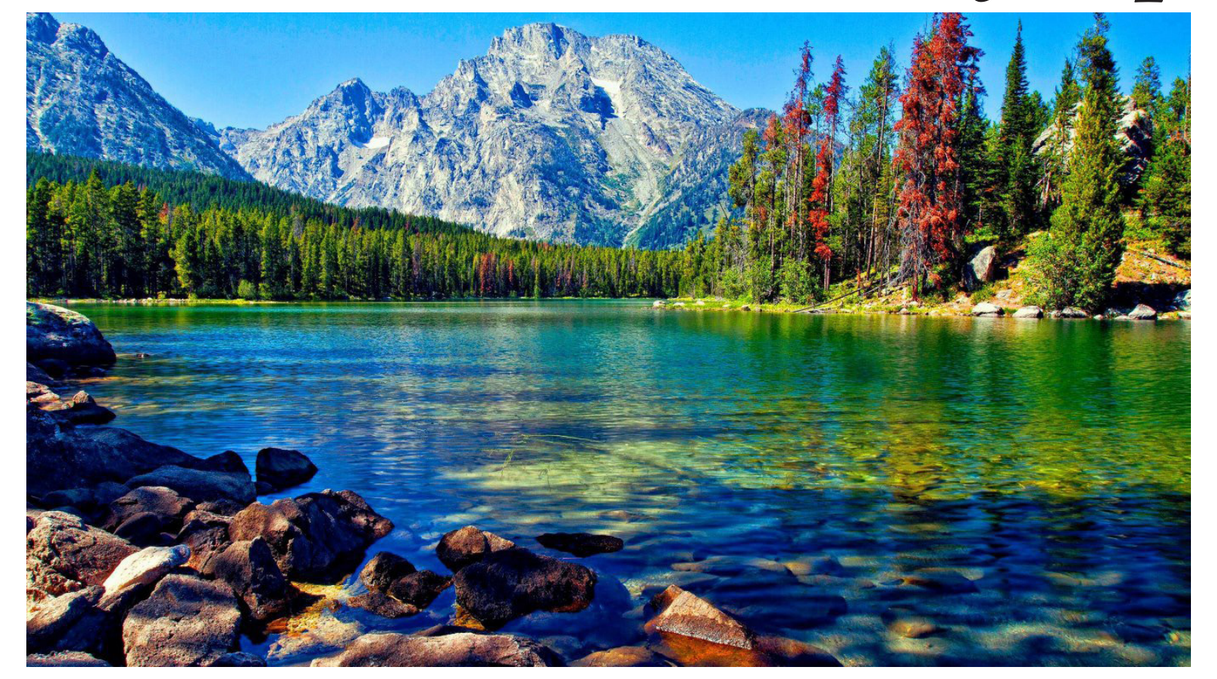

Images picked by the participant before the experiment began using the instructions, "Pick images that are **movingly beautiful to you.**"

Pretty IAPS

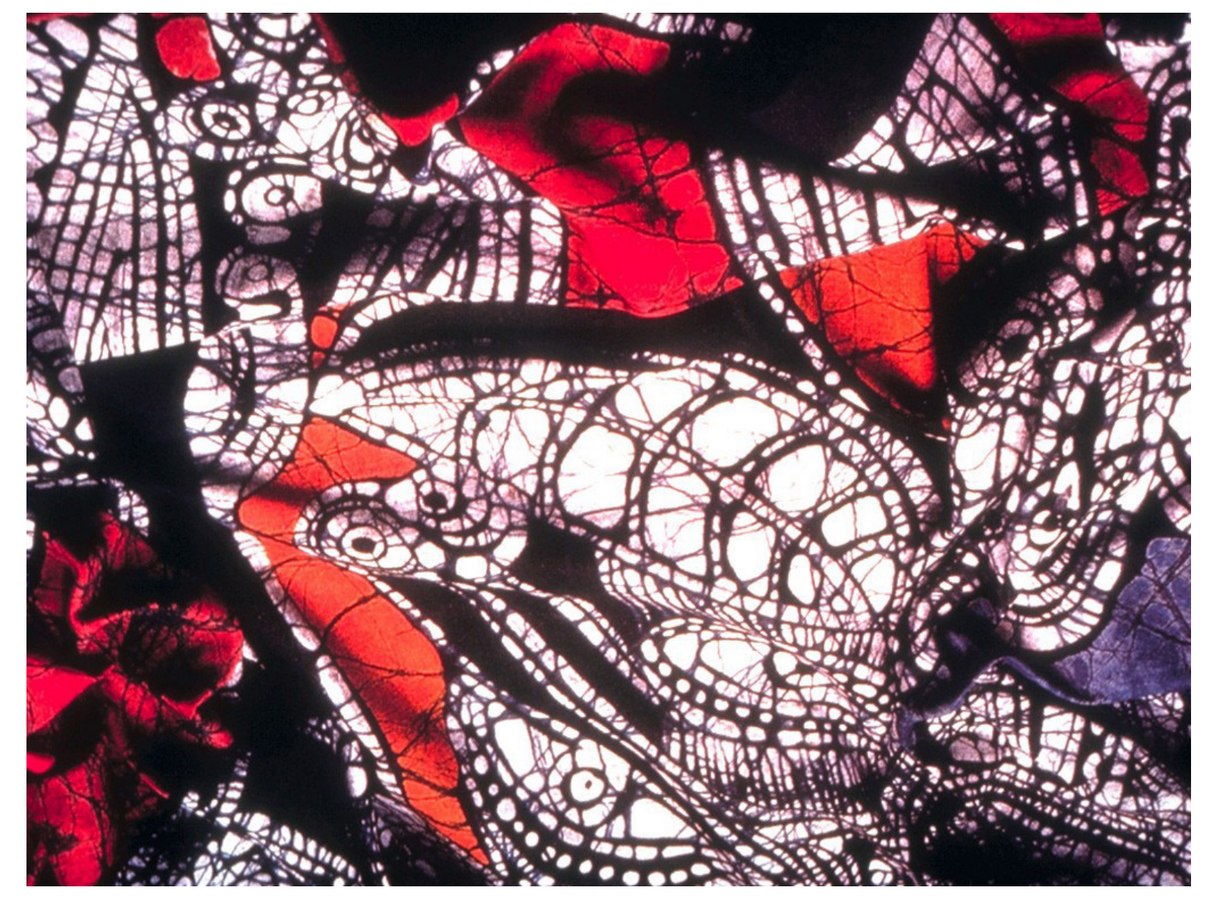

Images from IAPS [3] database previously rated medium on beauty, liking, and pleasantness.

IKEA neutral

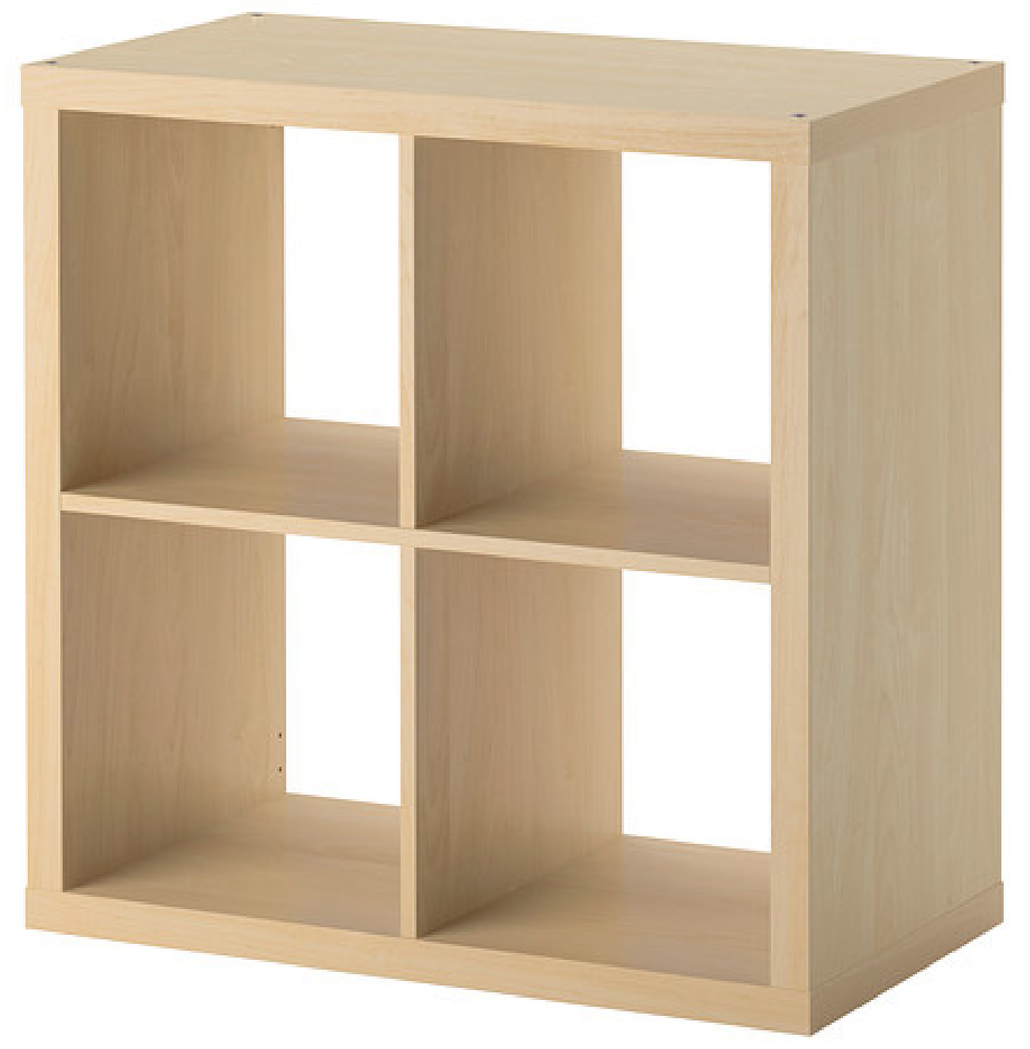

Images depicting simple furniture from an IKEA catalogue.

Beautiful IAPS

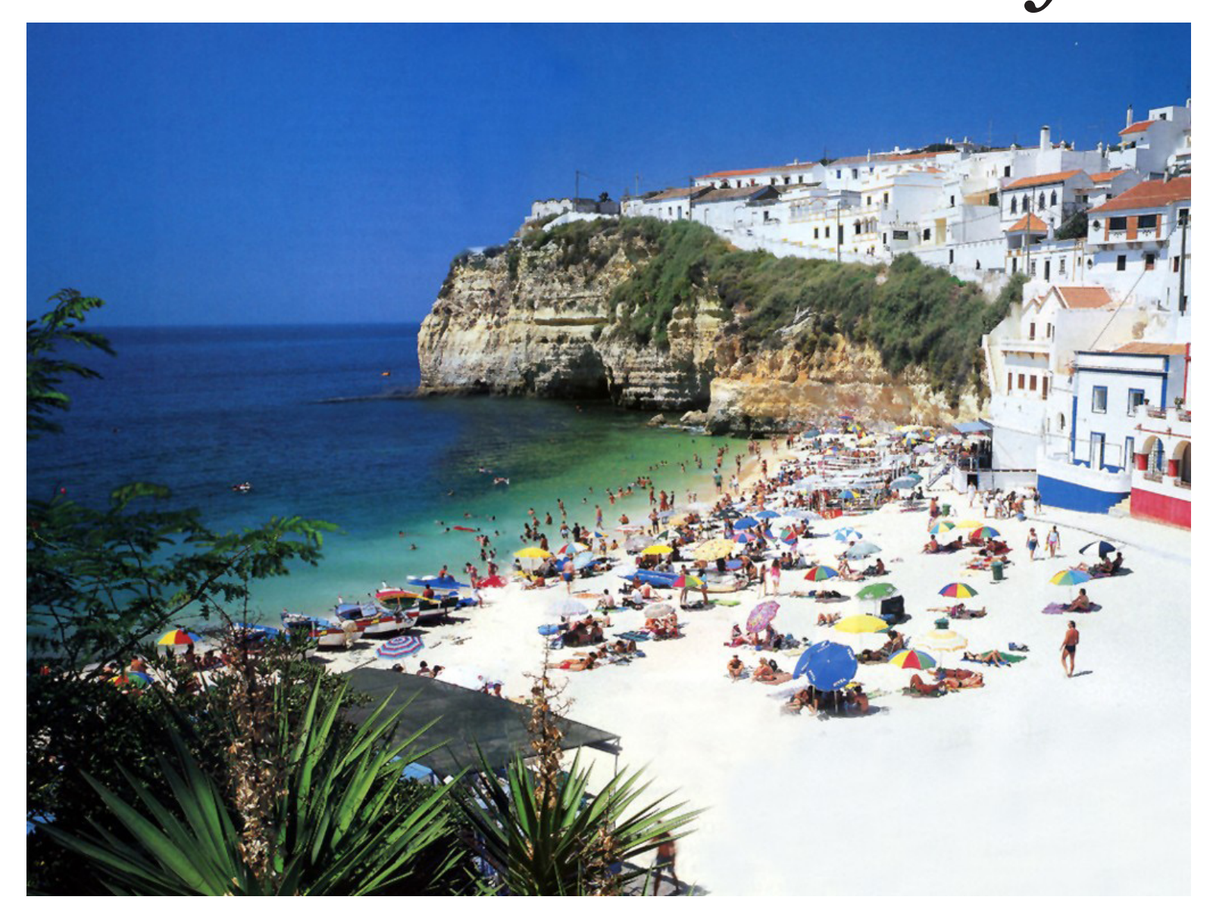

Images from IAPS [3] database previously rated high on beauty, liking, and pleasantness.

Candy

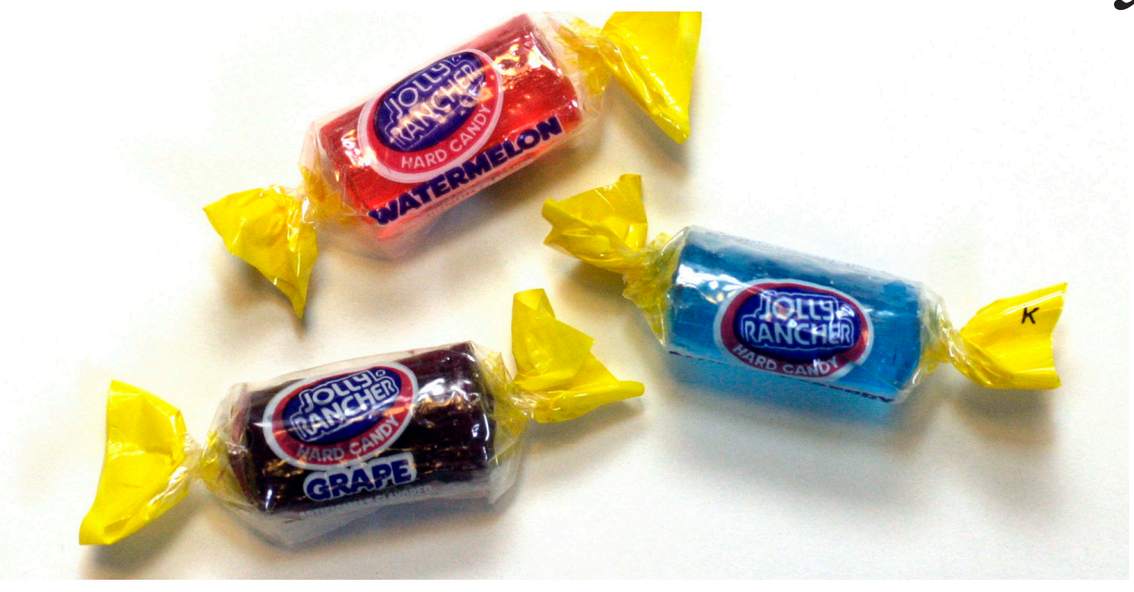

Jolly rancher hard candy.

Measurement

Each participant continuously reported pleasure by dynamically adjusting the spread of two fingers on a smartphone running our web app, EmotionTracker.com.

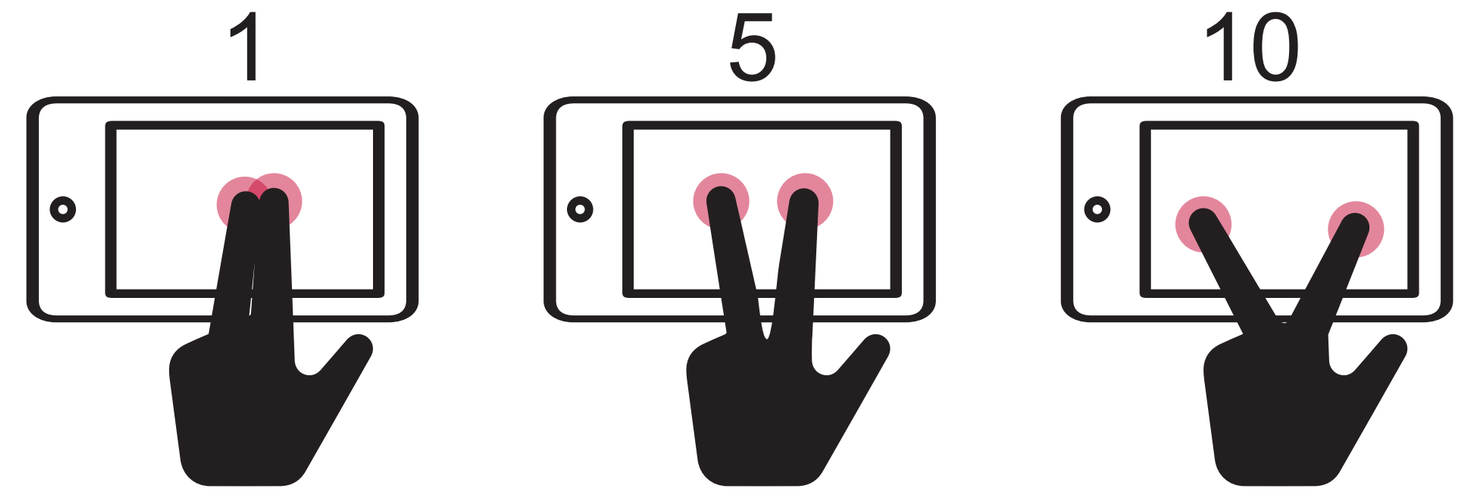
1
5
10

EmotionTracker.com

Trial sequence

Each *trial* presents a stimulus for 1, 6, 15, or 30 s, followed by a blank screen for 60 s. There were 20 trials presented in random order. Participants were asked to continuously rate their pleasure from the image for the entire trial.

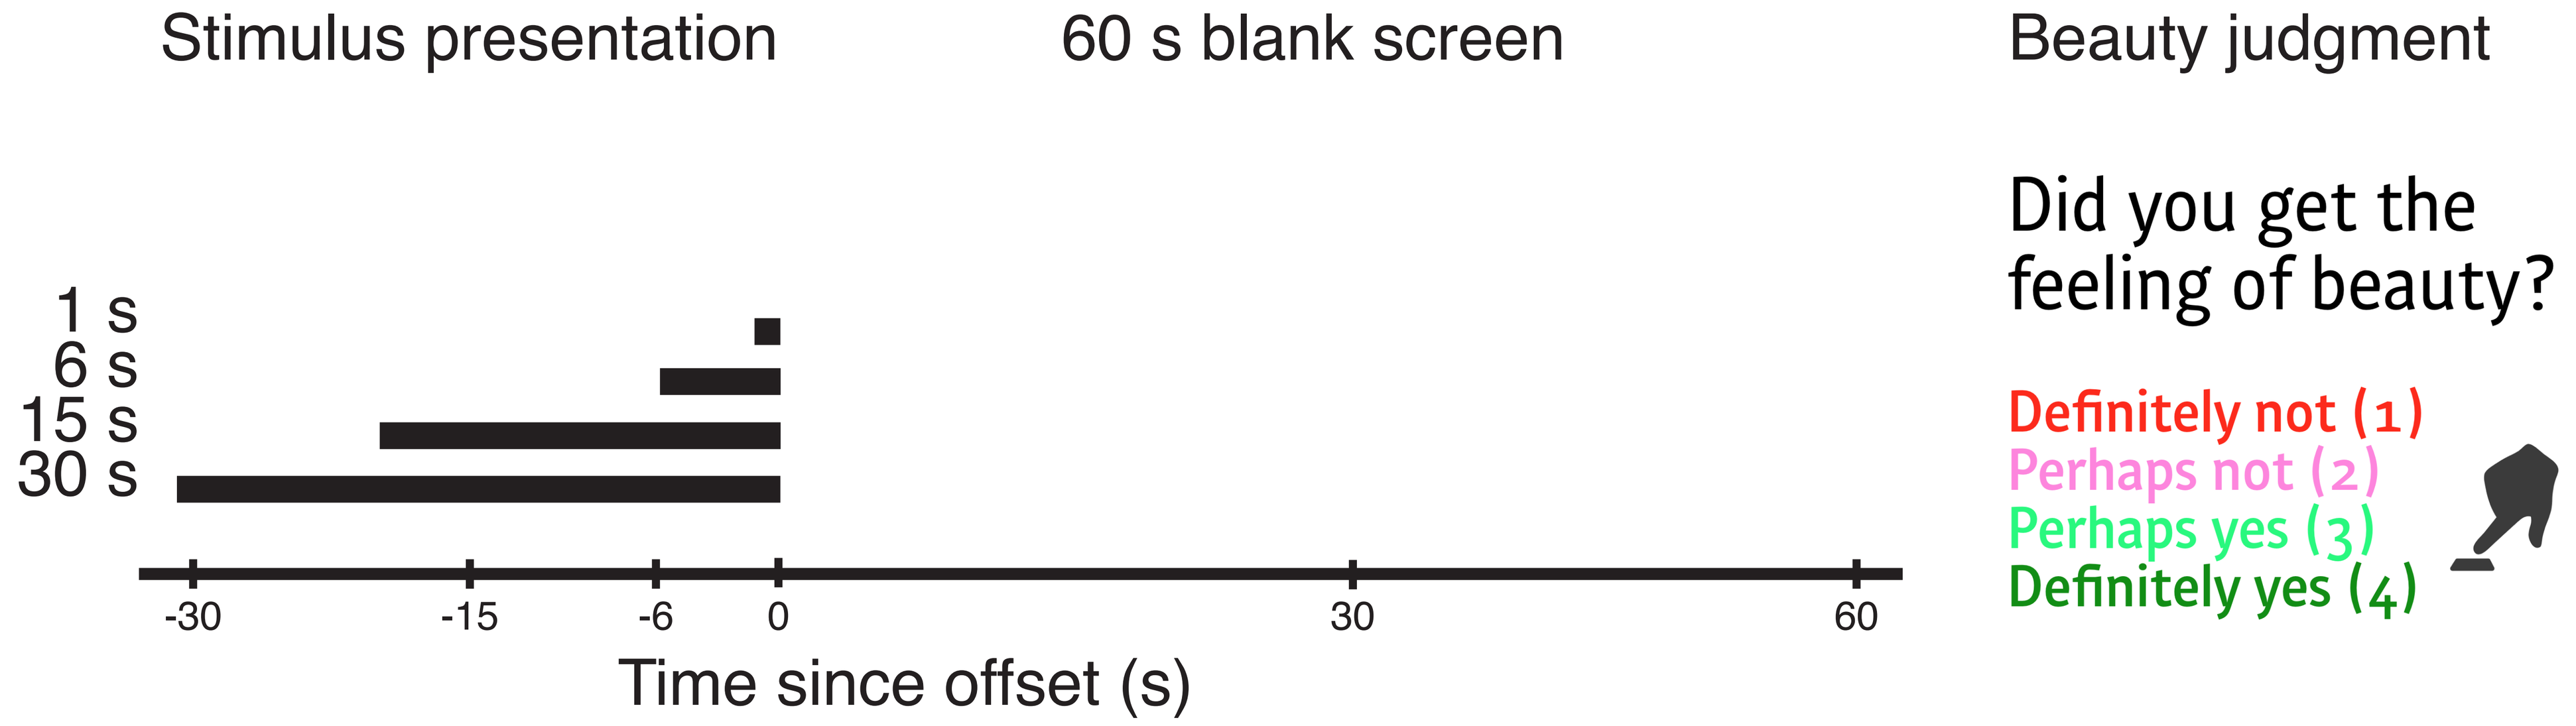
Stimulus presentation
60 s blank screen
Beauty judgment
Did you get the feeling of beauty?
Definitely not (1)
Perhaps not (2)
Perhaps yes (3)
Definitely yes (4)
1 s
6 s
15 s
30 s
-30
-15
-6
0
30
60
Time since offset (s)

Time course of self-reported pleasure

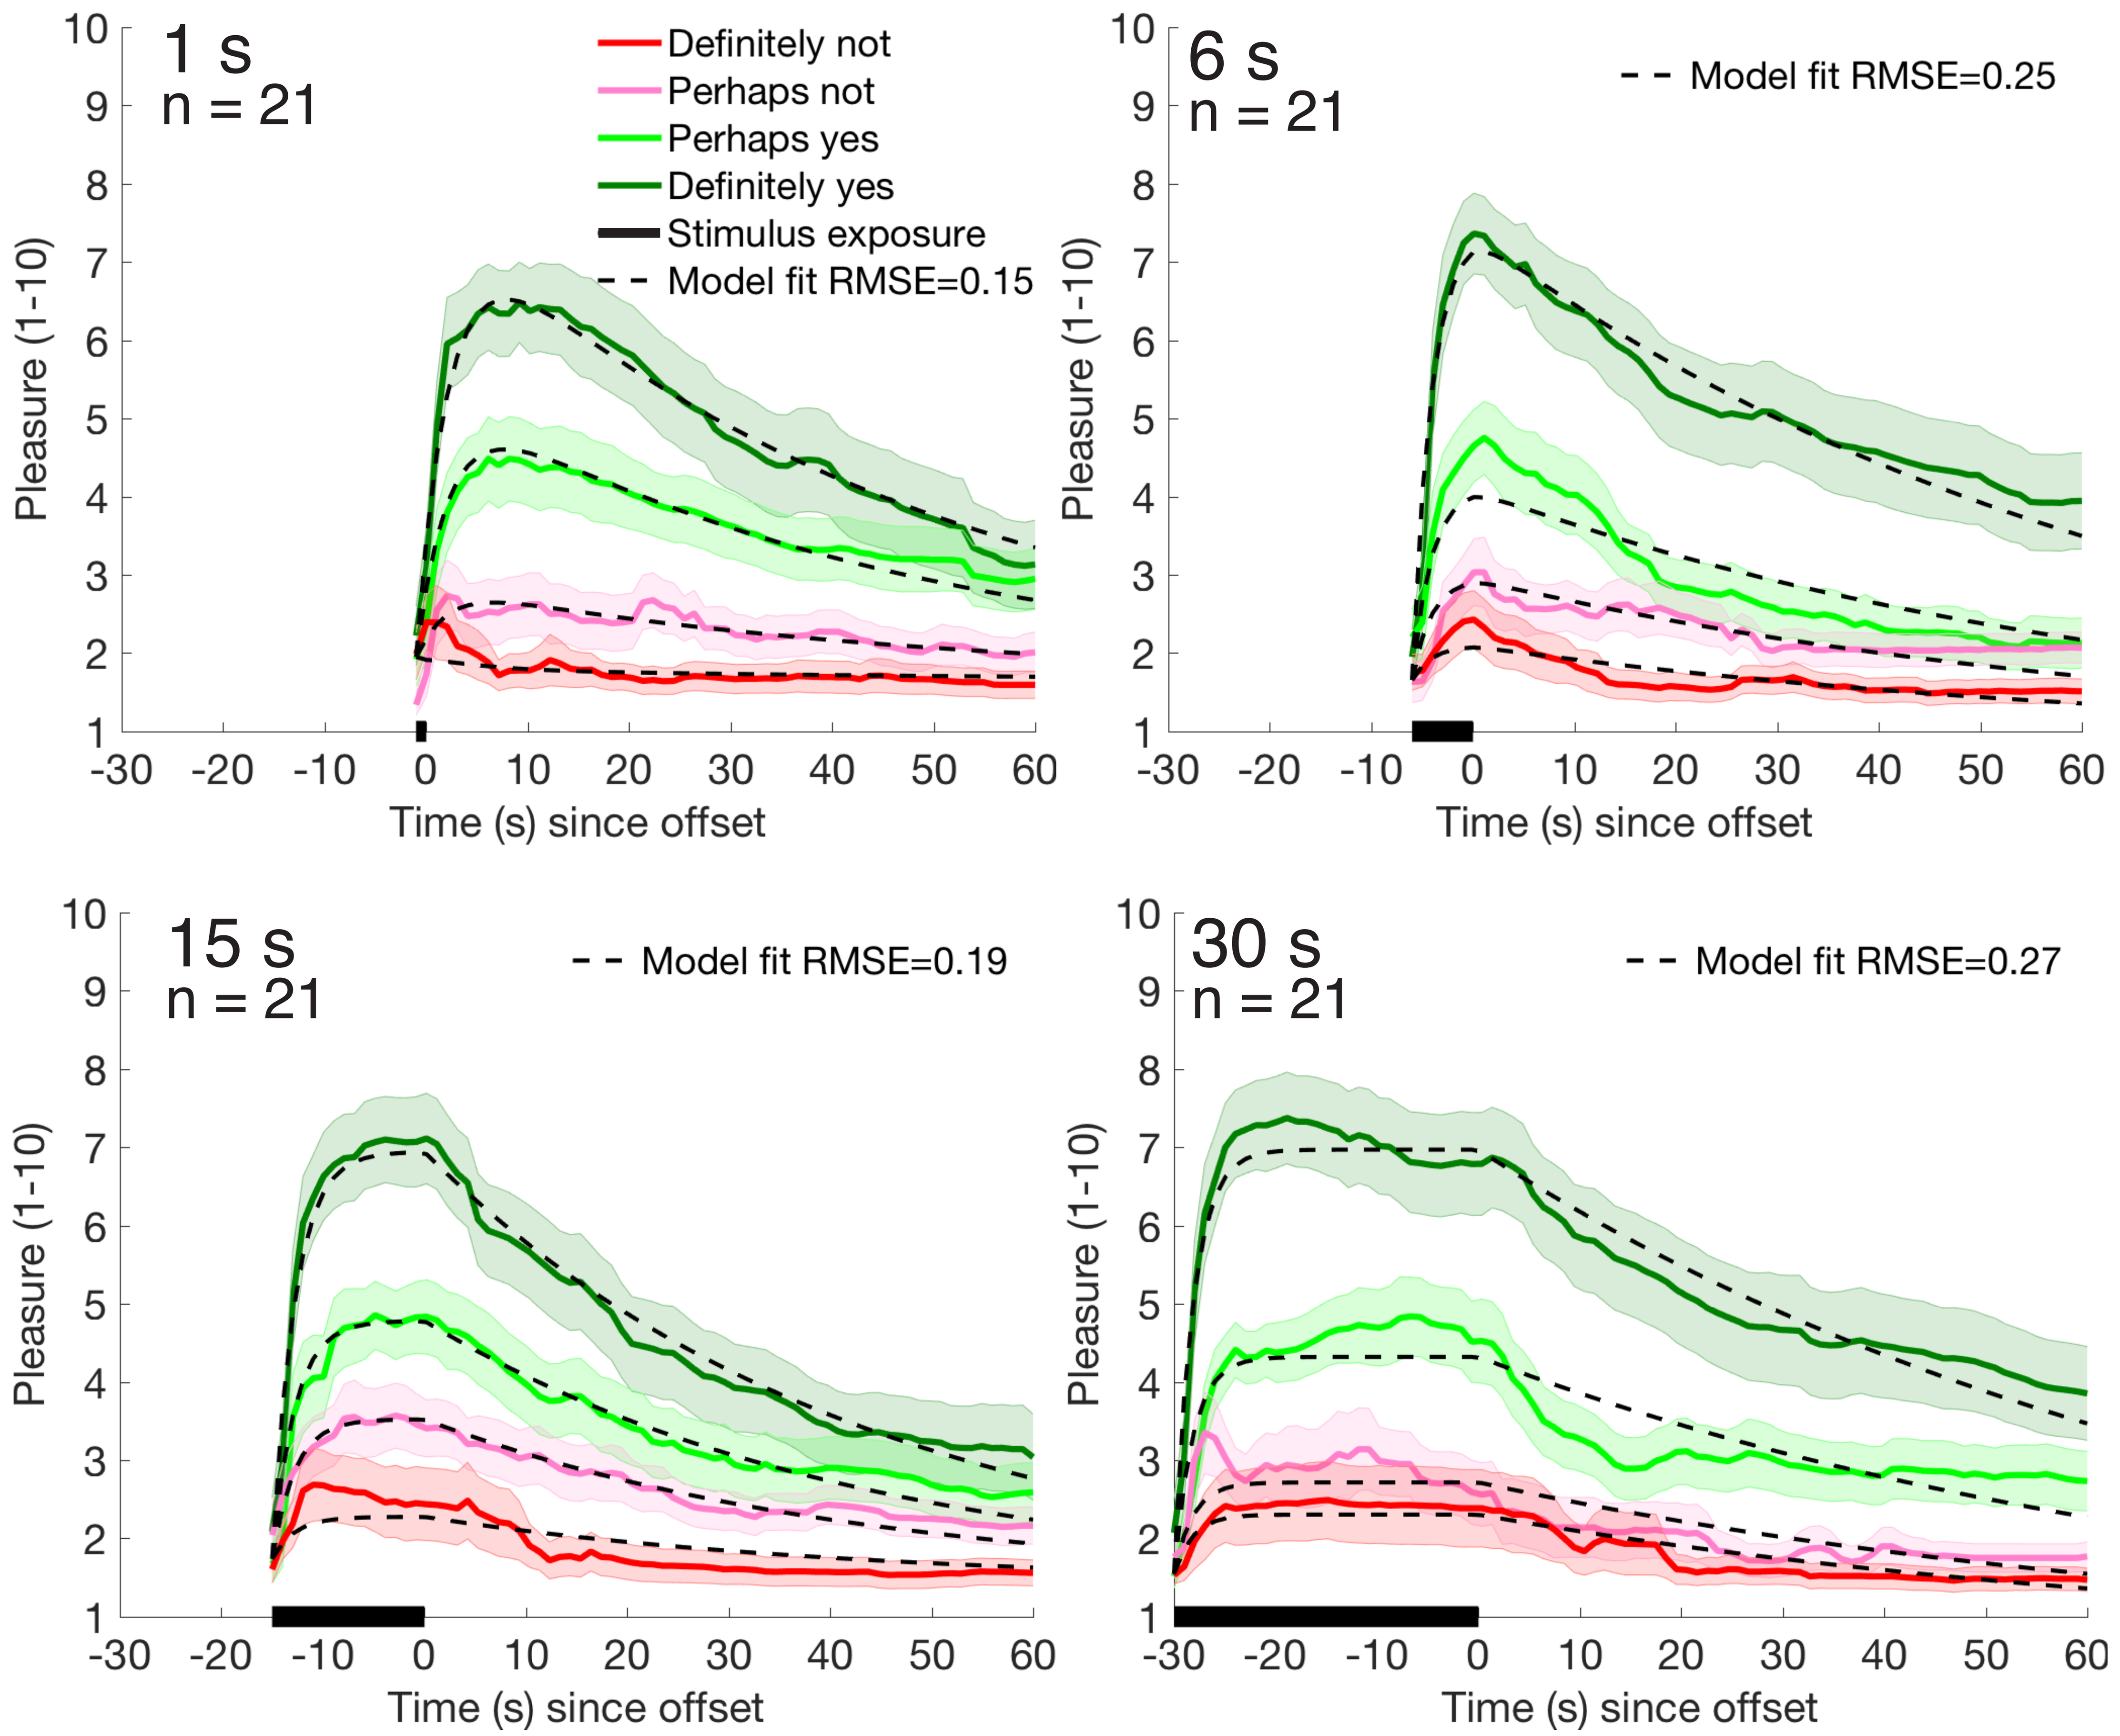
1 s
n = 21
6 s
n = 21
15 s
n = 21
30 s
n = 21
Definitely not
Perhaps not
Perhaps yes
Definitely yes
Stimulus exposure
Model fit RMSE=0.15
Model fit RMSE=0.25
Model fit RMSE=0.19
Model fit RMSE=0.27
Pleasure (1-10)
Time (s) since offset

Figure 1. Average pleasure over time for 1, 6, 15, and 30 s exposures. For each final beauty judgment, the data are averaged across participants. The shaded area indicates ±1 standard error of the mean. The black dashed lines indicate the model fit. Across all durations of exposure, pleasure is highest when participants reported definitely feeling beauty, and lowest when participants reported definitely not feeling beauty.

References

1. Brieber, D., Nadal, M., Leder, H., & Rosenberg, R. (2014). Art in time and space: context modulates the relation between art experience and viewing time. PloS one, 9(6), e99019.
2. Smith, J. K., & Smith, L. F. (2001). Spending time on art. Empirical Studies of the Arts, 19, 229–236.
3. Lang, P. J., Bradley, M. M., & Cuthbert, B. N. (1999). International affective picture system (IAPS): Technical manual and affective ratings.

Acknowledgements

We thank Stephanie Jose and Kimberly Wang for their help with data collection, Aenne Brielmann, Amy Belfi, Gwyneth Lewis, and Gizem Küçükoğlu helpful comments on the poster, Aenne Brielmann for the EmotionTracker.com graphics, and Gabrielle Starr, Ed Vessel, and the entire GIAS research group for their fruitful discussions. This work was funded by the NYU "Beauty and Beyond" GIAS research initiative.

Citation

Vale, L., & Pelli, D. G. (2016) The role of exposure duration in the experience of beauty. Vision Sciences Society, St. Petes Beach, Florida, May 13-18, 2016. http://psych.nyu.edu/pelli/posters.html

Modeling the time course

Inspection of the results led us to suppose a simple model with exponential transitions among three stable levels: before, steady state, and long after the stimulus. For each stimulus duration, our model supposes an initial response level r_{start} and a final asymptotic level r_{end} that are independent of the beauty judgment. After stimulus onset, the pleasure exponentially approaches the steady-state response r_{steady}, which does depends on the beauty judgment. After stimulus offset, pleasure decays exponentially towards the final level r_{end}. There are separate time constants τ_{start} and τ_{end} for onset and offset.

$$\hat{R}(t)=\left(1-\alpha_{start}(t)\right)r_{start}+\alpha_{start}(t)\left[\left(1-\alpha_{end}(t)\right)r_{steady}+\alpha_{end}(t)r_{end}\right] \quad (1)$$

$$\alpha_{start}(t)=1-\exp\frac{-\max(t-t_{start},0)}{\tau_{start}} \quad (2)$$

$$\alpha_{end}(t)=1-\exp\frac{-\max(t-t_{end},0)}{\tau_{end}} \quad (3)$$

$$\varepsilon=\sqrt{\operatorname{mean}_t\left(R(t)-\hat{R}(t)\right)^2} \quad (4)$$

Model parameter		1 s	6s	15 s	30 s
Response start	r_{start}	2.0	1.7	1.7	1.5
Response end	r_{end}	1.7	0.9	1.4	0.6
Start time constant (s)	τ_{start}	3.4	1.9	2.2	1.8
End time constant (s)	τ_{end}	46.3	66.5	42.6	75.5
RMS error of fit	ε	0.15	0.25	0.19	0.27

Table 1. Parameter estimates from the model fit for each duration of exposure. The steady state response stays consistent across exposure duration. The fits are good: RMSE < 0.3 on our 10-point pleasure scale.

Steady-state pleasure vs. beauty

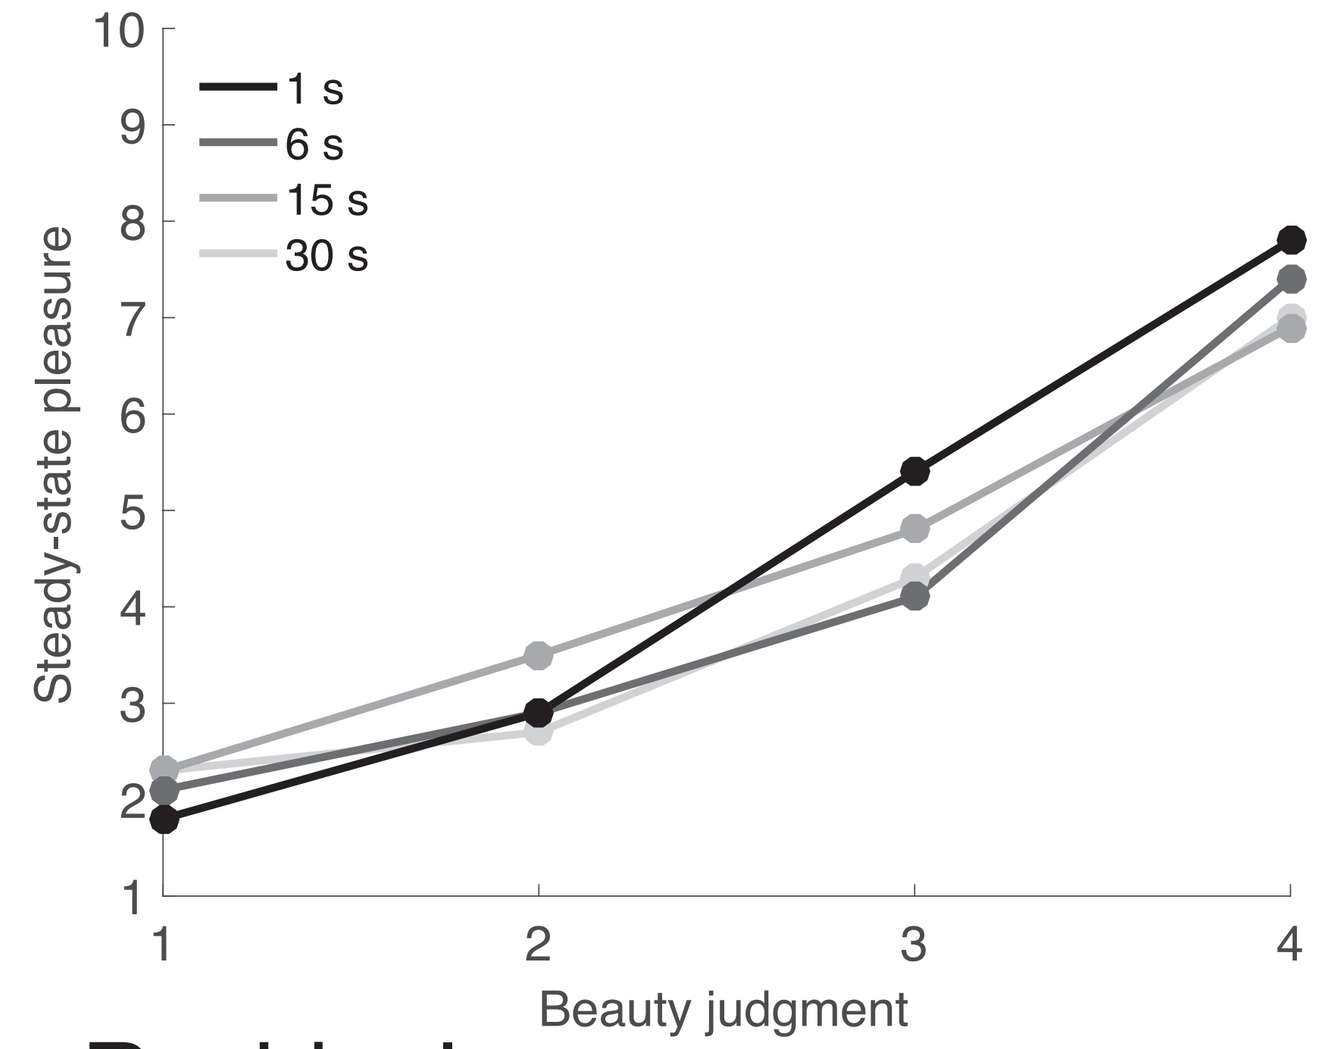
1 s
6 s
15 s
30 s
Steady-state pleasure
Beauty judgment

Figure 2. Steady-state pleasure r_{steady} vs. beauty judgement. Pleasure grows roughly linearly with the judged beauty.

Steady-state pleasure vs. duration

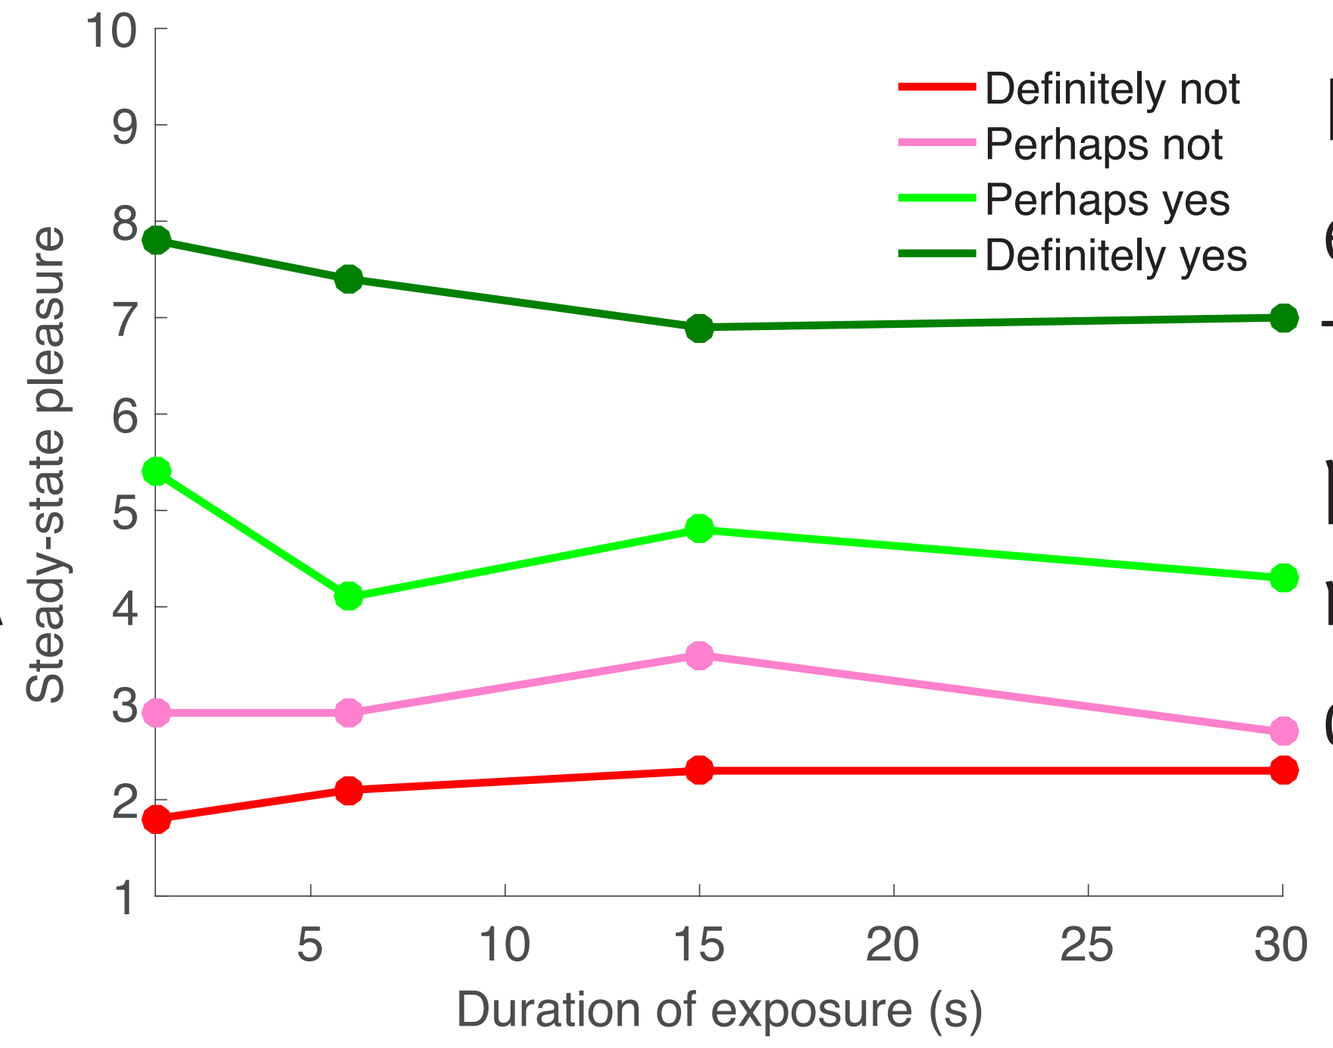
Definitely not
Perhaps not
Perhaps yes
Definitely yes
Steady-state pleasure
Duration of exposure (s)

Figure 3. The effect of duration. The steady-state pleasure estimate is roughly independent of duration.

Residuals

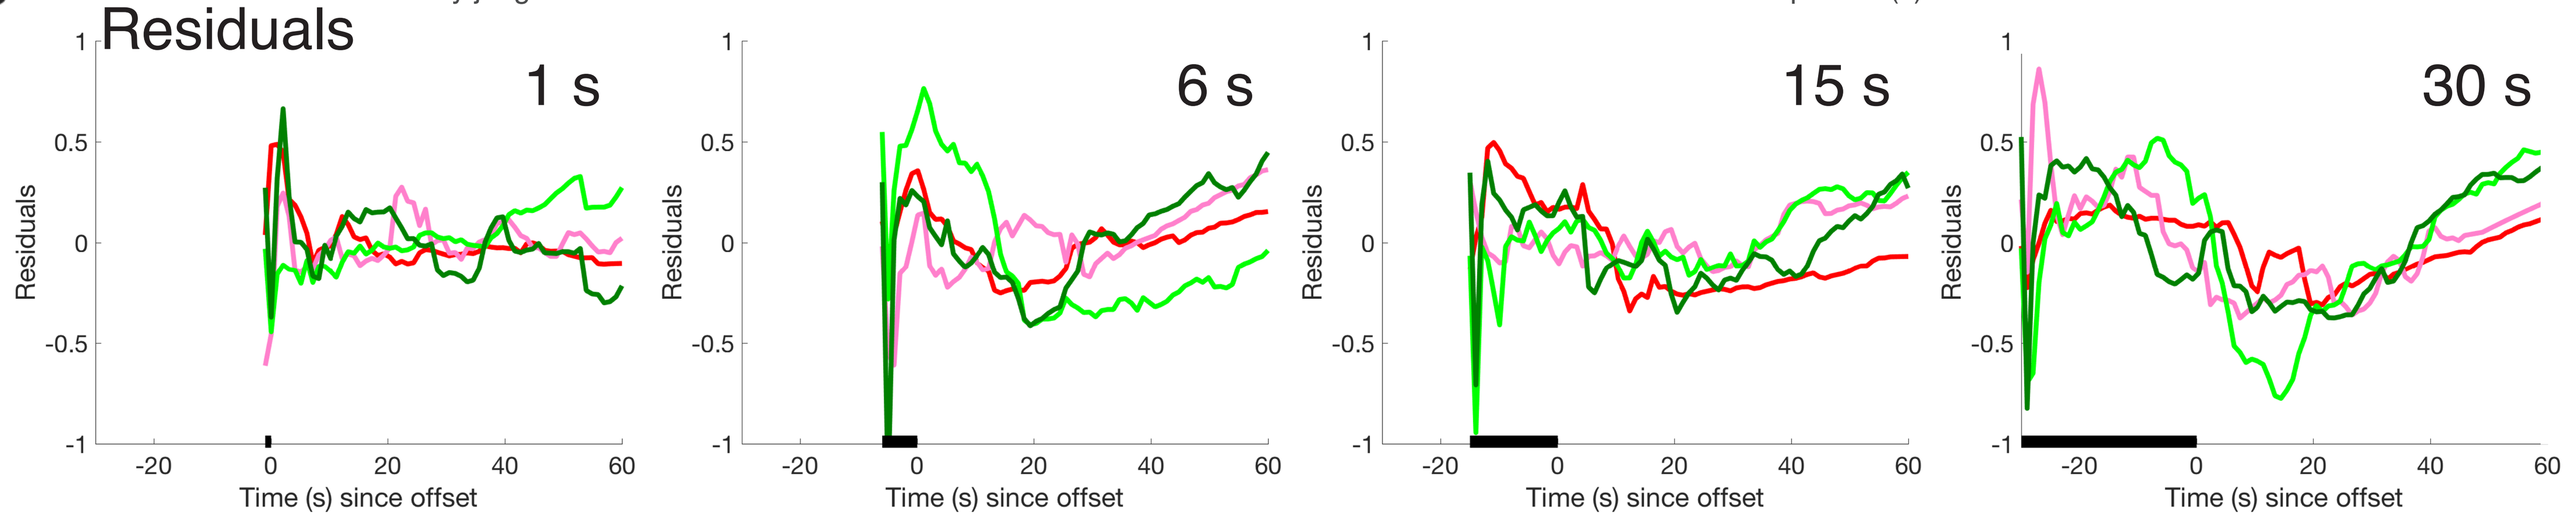
1 s
6 s
15 s
30 s
Residuals
Time (s) since offset

Figure 4. Residuals from the model fits. The residual shows a slow shallow U-shaped droop and rise over the course of the whole trial, independent of the beauty judgement.

Conclusion

A very simple model fits all our results surprisingly well. (The RMS error in pleasure is less than 0.3 points on our 10-point scale.) To a good approximation, steady-state pleasure grows linearly with the beauty judgement, independent of duration. The rest of the model parameters are conserved across duration & beauty judgement. Onset has a time constant of roughly 2 s, and offset has a time constant of roughly 60 s.